Poster 2621P
Abstract #3049

Central Nervous System (CNS) Activity of Zipalertinib in Non-Small Cell Lung Cancer (NSCLC) Patients With Uncommon *EGFR* Mutations (*EGFR*mt)

Helena A. Yu,[1] Kadoaki Ohashi,[2] Makoto Nishio,[3] Jon Zugazagoitia,[4] Raffaele Califano,[5] Maurice Perol,[6] Yasir Elamin,[7] Dae Ho Lee,[8] Jose Antonio López,[9] Alexander Spira,[10] Hiroshi Tanaka,[11] Santiago Viteri,[12] Muriel Granier,[13] Li Wei,[13] Maciej Gil,[13] Javier De Castro[14]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Okayama University Hospital, Okayama, Japan; [3]The Cancer Institute Hospital, Japanese Foundation for Cancer Research, Tokyo, Japan; [4]Hospital Universitario 12 de Octubre, Madrid, Spain; [5]The Christie NHS Foundation Trust and Division of Cancer Sciences, The University of Manchester, Manchester, UK; [6]Centre Leon Berard, Lyon, France; [7]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [8]University of Ulsan College of Medicine, Asan Medical Center, Seoul, Korea; [9]Hospital Universitario de Jaén, Jaén, Spain; [10]Virginia Cancer Specialists, Fairfax, VA, USA; [11]Niigata Cancer Center Hospital, Niigata, Japan; [12]UOMI Cancer Center, Clinica Mi Tres Torres, Barcelona, Spain; [13]Taiho Oncology, Inc., Princeton, NJ, USA; [14]Hospital Universitario La Paz, Madrid, Spain

Introduction

- CNS metastases in patients with NSCLC harboring *EGFR* mutations are associated with poor prognosis[1]
- There are limited treatment options for patients with NSCLC harboring *EGFR* ex20ins mutations with CNS metastases[2]
- Zipalertinib is an oral, highly selective, irreversible EGFR tyrosine kinase inhibitor (TKI) that has demonstrated clinical activity against advanced/metastatic NSCLC harboring *EGFR* ex20ins mutations in a phase 1/2 trial, including in patients with CNS metastases[3–5]
- Zipalertinib also demonstrated clinical activity in patients with locally advanced/metastatic NSCLC harboring other uncommon mutations, including L861Q, G719X, S768I, and compound mutations[6]
- Here we report follow-up results from Cohort C of the ongoing phase 2b REZILIENT2 trial (NCT05967689), focusing on patients with NSCLC harboring *EGFR* ex20ins or other uncommon mutations (including L861Q, G719X, S768I) and compound mutations, who have active newly diagnosed and/or progressing brain metastases and/or leptomeningeal disease (LMD)

Methods: Study Design

REZILIENT2 is a multicohort open-label, phase 2 trial (NCT05967689; EUDRA CT 2023-503865-48)

Eligibility
- Age ≥18 years with locally advanced/metastatic NSCLC
- Documented **ex20ins or other uncommon single or compound non-ex20ins *EGFR* mutations[a]**
- Measurable disease per **RECIST v1.1** and CNS metastases by **RANO-BM**
- **Active brain metastases**: newly diagnosed and/or progressing brain lesions (without CNS targeted therapy) and/or LMD
- ECOG PS 0 or 1
- Any prior line of therapy for advanced/metastatic disease

Endpoints (investigator assessed)

Primary:
- **ORR** per RECIST v1.1

Secondary:
- Intracranial ORR, intracranial DOR, and intracranial DCR per RANO-BM
- Safety

[a]Determined by local testing performed at a CLIA certified (US) or locally certified laboratory (outside the US).
CNS, central nervous system; DCR, disease control rate (complete response + partial response + stable disease); DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group Performance Status; EGFR, epidermal growth factor receptor; ex20ins, exon 20 insertion; LMD, leptomeningeal disease; NSCLC, non-small cell lung cancer; ORR, objective response rate (complete response + partial response); RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; RECIST, Response Evaluation Criteria in Solid Tumors.

- Data cutoff: June 27, 2026; study is ongoing
- Patients received zipalertinib at a starting dose of 100 mg orally twice daily until progressive disease or other withdrawal criteria
- Median number of cycles received was 7 (range, 1–35)

Results: Patients

- Of 59 enrolled patients, 43 were Response Assessment in Neuro-Oncology Brain Metastases (RANO-BM) measurable (**Figure 1**)

Figure 1. Patient disposition

Cohort C
Patients enrolled
N=59

RECIST v1.1 measurable
n=59

RANO-BM measurable
n=43

Discontinued treatment n=51
AE/SAE: n=12
Clinical disease progression: n=6
Radiographic disease progression[a]: n=28
Withdrew consent: n=1
Death: n=4

Discontinued treatment n=37
AE/SAE: n=10
Clinical disease progression: n=3
Radiographic disease progression[a]: n=19
Withdrew consent: n=1
Death: n=4

Treatment ongoing at data cutoff[b]
n=8

Treatment ongoing at data cutoff[b]
n=6

[a]Disease progression as per RECIST v1.1; [b]Data cutoff: June 27, 2026.
AE, adverse event; RANO-BM, Response Assessment in Neuro-oncology Brain Metastases; RECIST, Response Evaluation Criteria in Solid Tumors; SAE, serious adverse event.

- Median age of patients was 62 years (range, 23–84) in both groups and the majority had ex20ins mutations (**Table 1**)
- 13/59 patients (22%) had LMD (**Table 1**)
- 27/43 RANO-BM measurable patients (63%) and 41/59 of all enrolled patients (69%) had not received prior CNS radiotherapy (**Table 2**)

Table 1. Patient demographics and disease characteristics

	Cohort C Total (N=59)	RANO-BM Measurable (n=43)
Median age (range), years	62 (23–84)	62 (23–84)
Female, n (%)	34 (58)	24 (56)
Race, n (%)		
White	30 (51)	24 (56)
Asian	21 (36)	13 (30)
Black or African American	1 (2)	1 (2)
Not collected	7 (12)	5 (12)
ECOG performance status 0/1	15 (25)/44 (75)	12 (28)/31 (72)
EGFR exon20 mutations, n (%)	40 (68)[a]	29 (67)[a]
A767_V769dup (ASV)	11 (19)	9 (21)
S768_D770dup (SVD)	4 (7)	3 (7)
N771_H773dup (NPH)	2 (3)	1 (2)
H773dup (H)	3 (5)	3 (7)
Other	22 (37)	15 (35)
Other EGFR mutations, n (%)	20 (34)	15 (35)
G719X	4 (7)	4 (9)
S768I	1 (2)	1 (2)
L861Q	3 (5)	1 (2)
E709X	1 (2)	1 (2)
Compound mutations	9 (15)	7 (16)
Other[b]	2 (3)	1 (2)
Leptomeningeal disease, n (%)	13 (22)	9 (21)

[a]One patient had more than one mutation. [b]Other EGFR mutations were G779F (n=1) and I740_K745dup (n=1).
ECOG, Eastern Cooperative Group; EGFR, epidermal growth factor receptor; RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases.

Table 2. Prior therapies

	Cohort C Total (N=59)	RANO-BM Measurable (n=43)
Patients with at least 1 prior systemic drug therapy, n (%)	46 (78)	32 (74)
Median number of prior therapies (range)[a]	2 (1–6)	2 (1–6)
Prior systemic therapies, n (%)		
0	13 (22)	11 (26)
1	16 (27)	11 (26)
2	13 (22)	9 (21)
≥3	17 (29)	12 (30)
Median number of prior lines of EGFR-TKI (range)[b]	1 (1–3)	1 (1–2)
Prior lines of EGFR-TKI therapies, n (%)		
0	34 (58)	24 (56)
1	16 (27)	13 (30)
2	8 (14)	6 (14)
≥3	1 (2)	0
Prior amivantamab therapy, n (%)	9 (15)	8 (19)
Prior CNS radiotherapy, n (%)	18 (31)	16 (37)
Time from end of therapy to first zipalertinib dose, range, months	1–62	1–62

[a]RANO-BM: n=32; Total: N=46. [b]RANO-BM: n=19; Total: N=25.
CNS, central nervous system; EGFR-TKI, epidermal growth factor receptor–targeted tyrosine kinase inhibitor; RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases.

Results: Efficacy

At data cut-off
- For RANO-BM measurable patients (n=43), median time on treatment was 4.4 months (range, 0.3–21.4)
- 13/43 patients (ORR 30%; 95% CI: 56, 85) with measurable CNS disease had confirmed responses by RANO-BM criteria (including 2 intracranial complete response [CR]) (**Figures 2** and **3**)
 - Median intracranial duration of response (DOR) was 9.7 months (95% CI: 3.1, not evaluable [NE])
- Of the 13 patients with confirmed intracranial response
 - 10 patients had no prior CNS radiotherapy; the remaining 3 had prior CNS radiotherapy >8 months before the start of zipalertinib treatment
 - 3 patients also had LMD at baseline; median time on treatment was 6.8 months (range, 3.0–12.6)
- For RECIST measurable patients (N=59), median time on treatment was 4.4 months (range, 0.3–27.4)
- 18/59 patients (objective response rate [ORR] 31%; 95% CI: 19, 44) had confirmed systemic responses by RECIST v1.1 (**Figures 2** and **3**)
 - Median DOR was 8.4 months (95% CI: 4.4, 15.6)
- Long-term data show similar intracranial and systemic response rates

Figure 2. Responses

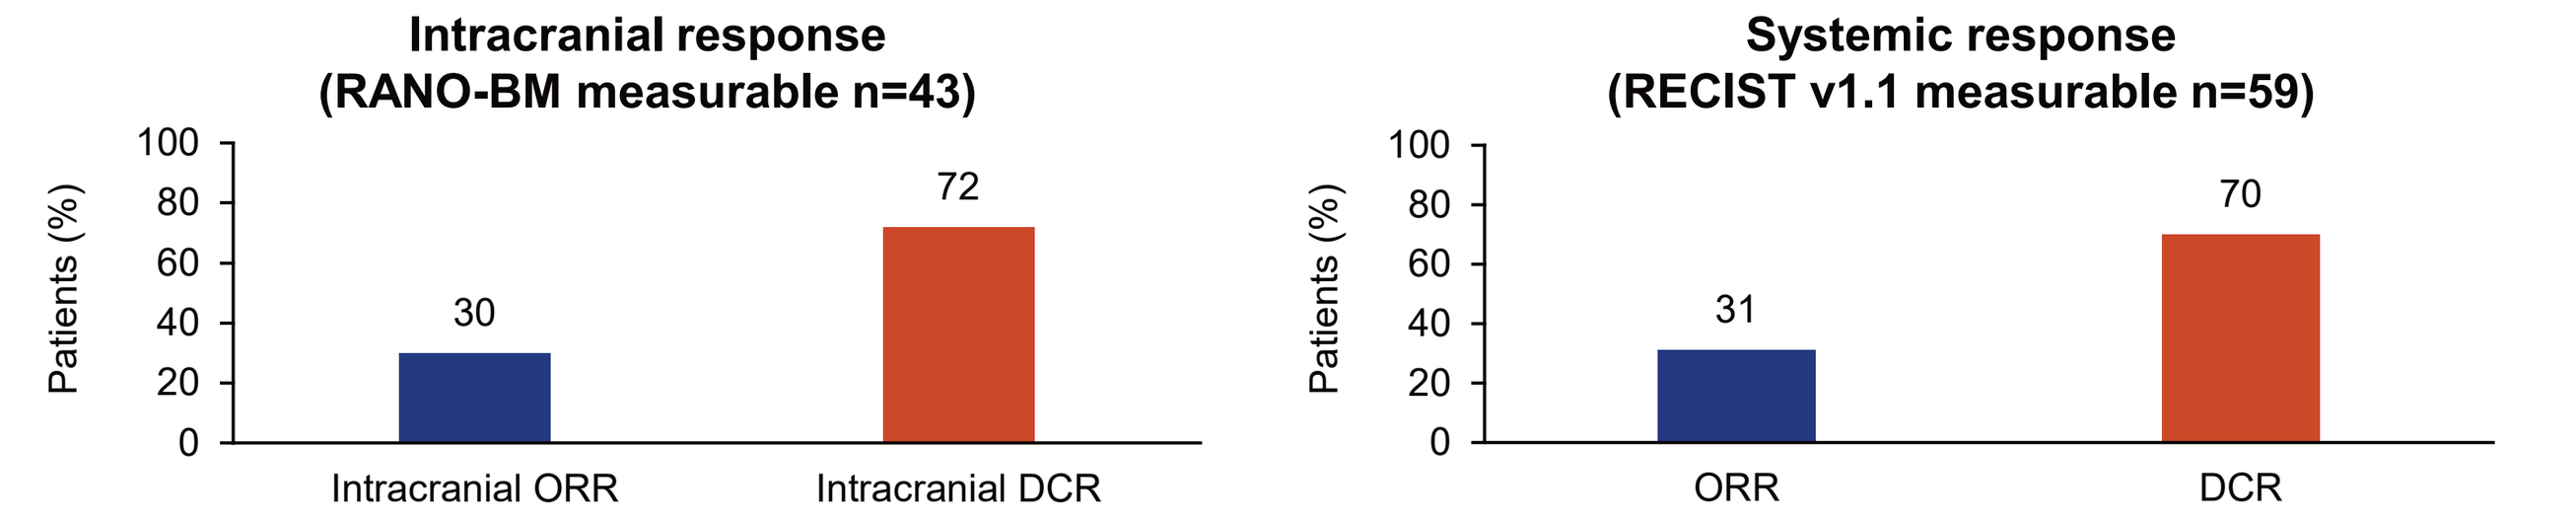

Intracranial response
(RANO-BM measurable n=43)
Patients (%)
100
80
60
40
20
0
30
72
Intracranial ORR
Intracranial DCR
Systemic response
(RECIST v1.1 measurable n=59)
31
70
ORR
DCR

DCR, disease control rate (complete response + partial response + stable disease); ORR, objective response rate (complete response + partial response); RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, Version 1.1.

Figure 3. Best percentage change in target lesions

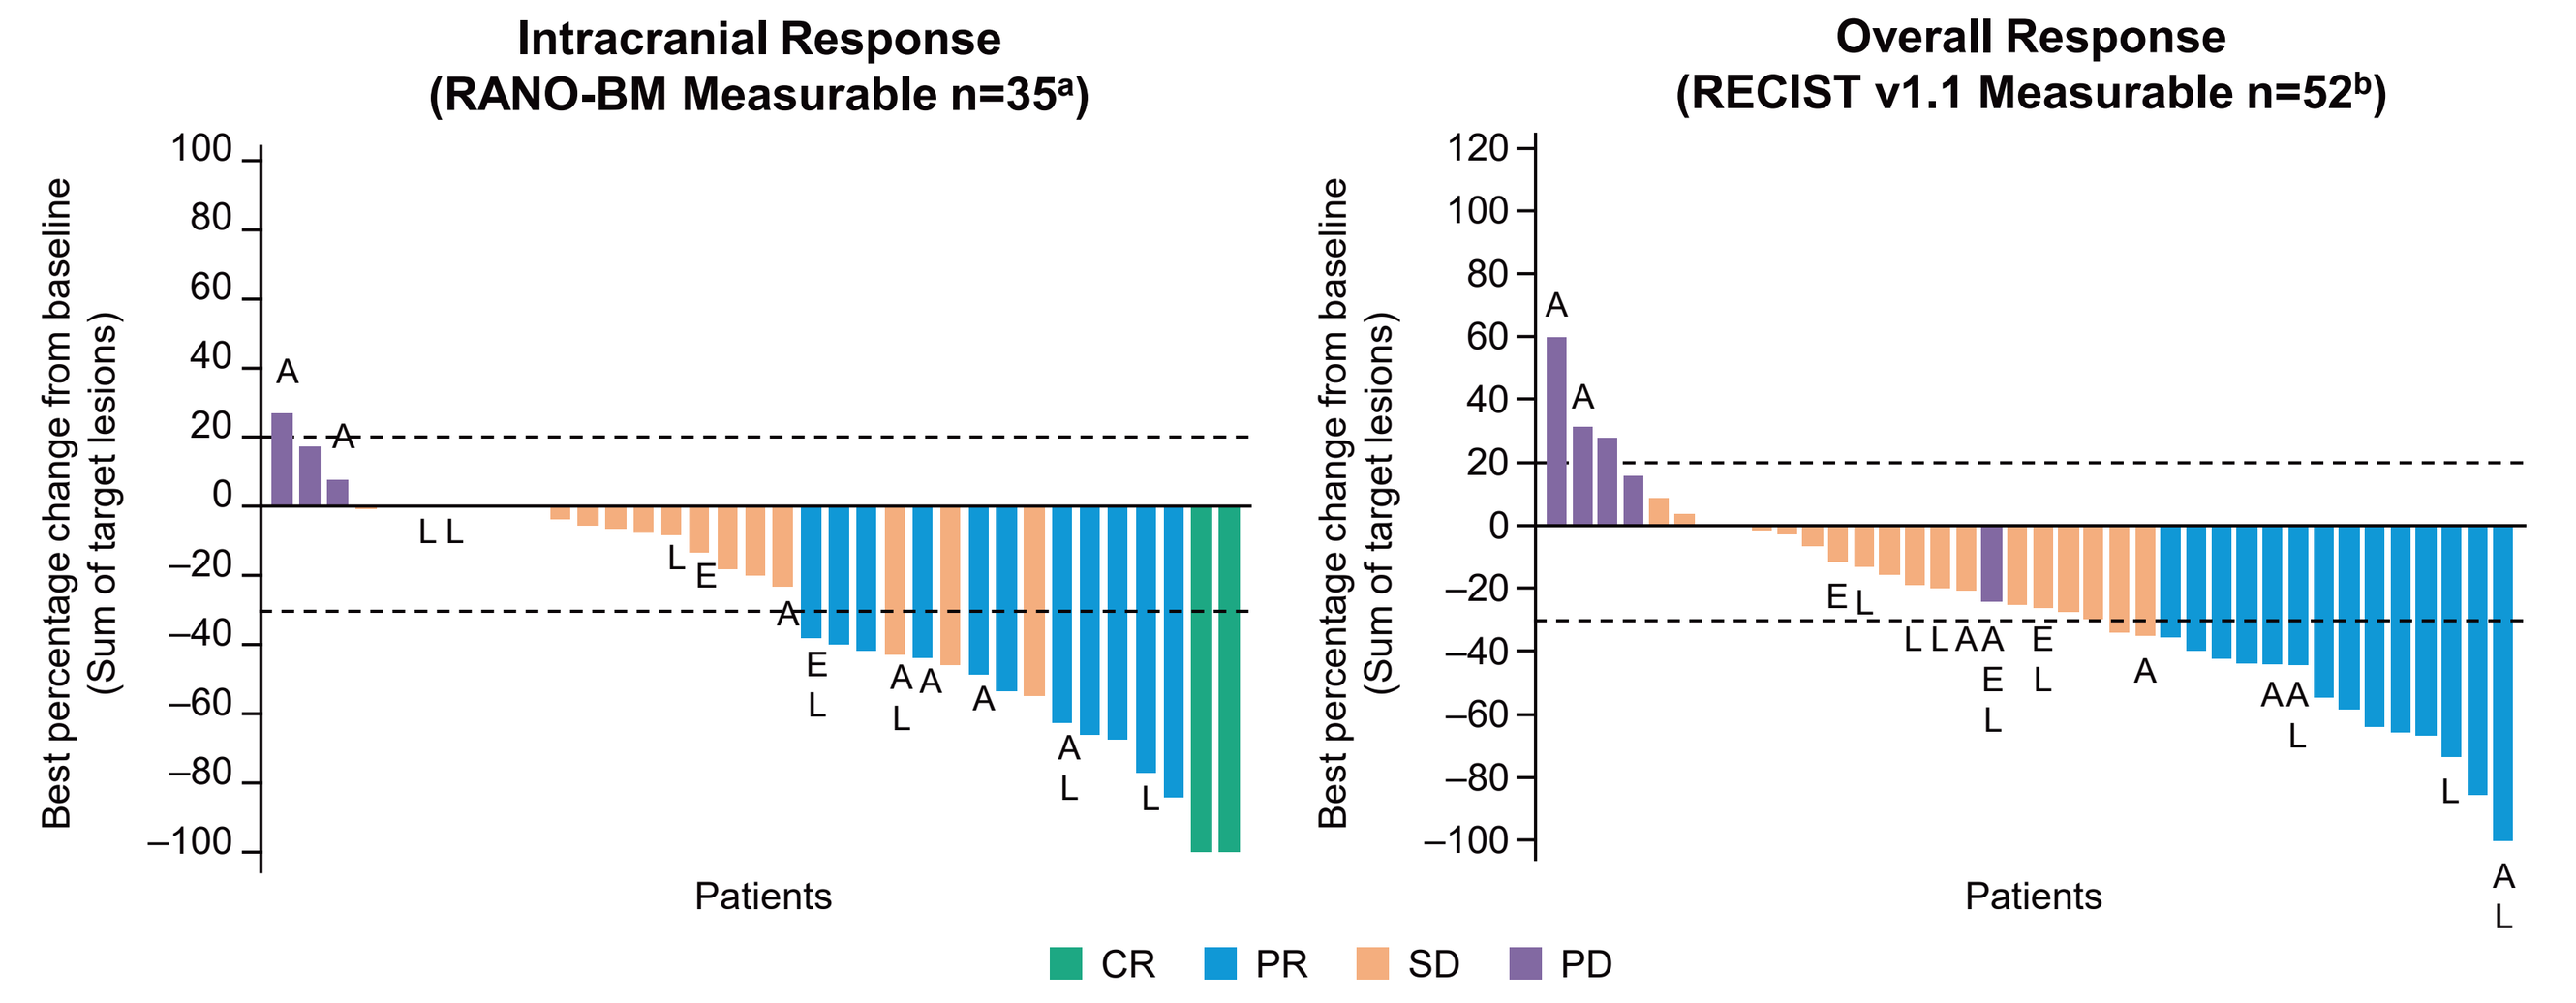

Intracranial Response
(RANO-BM Measurable n=35[a])
Overall Response
(RECIST v1.1 Measurable n=52[b])
Best percentage change from baseline
(Sum of target lesions)
Patients
CR
PR
SD
PD

[a]8 of 43 patients were not evaluable for response. [b]7 of 59 patients were not evaluable for response.
A, prior amivantamab therapy; CR, complete response; E, prior *EGFR* exon20 targeted therapy (other than amivantamab); L, leptomeningeal disease; PD, progressive disease; PR, partial response; RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, Version 1.1; SD, stable disease.

- **Figure 4** shows treatment duration and individual patient responses by RANO-BM criteria and RECIST v1.1
- Zipalertinib demonstrated comparable systemic and intracranial activity in ex20ins NSCLC with brain metastases (RECIST v1.1 ORR 37.5%; RANO-BM ORR 34.5%)
- Responses occurred after prior amivantamab-only treatment (RECIST v1.1 4/7; RANO-BM 3/6). Antitumor activity after prior ex20ins TKIs was limited (RECIST 0/4; RANO-BM 1/3)
- In non-ex20ins NSCLC, predominantly pretreated with EGFR TKIs, response rates were modest (RECIST v1.1 3/19; RANO-BM 3/14)

Figure 4. Characterization of response

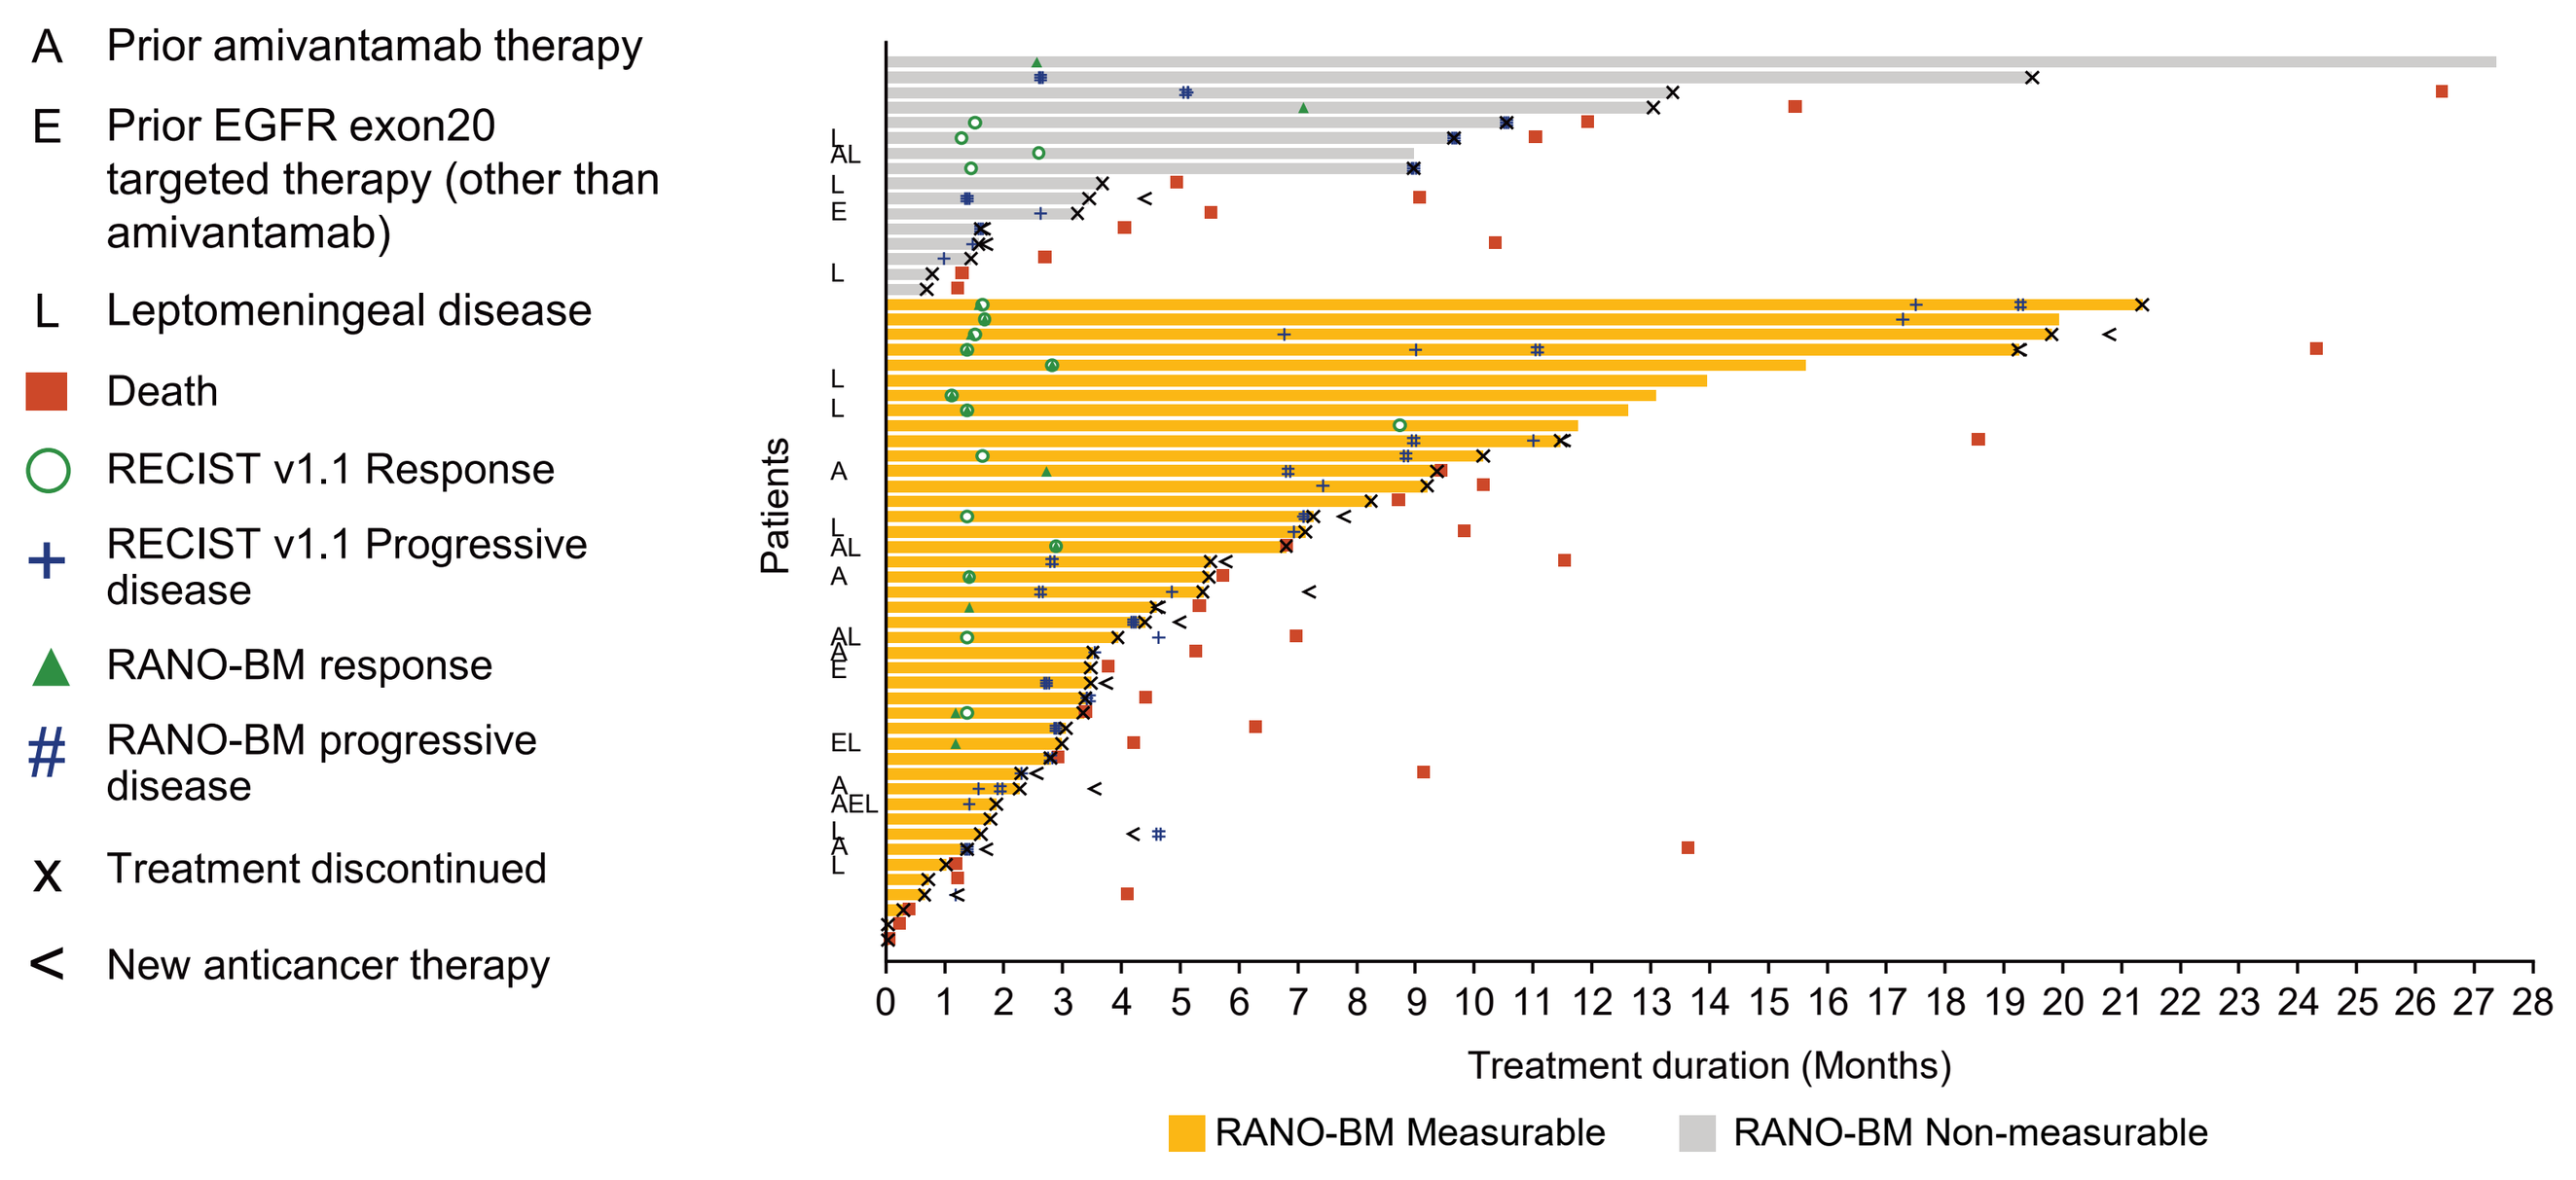

A Prior amivantamab therapy
E Prior EGFR exon20 targeted therapy (other than amivantamab)
L Leptomeningeal disease
Death
RECIST v1.1 Response
RECIST v1.1 Progressive disease
RANO-BM response
RANO-BM progressive disease
X Treatment discontinued
< New anticancer therapy
Patients
Treatment duration (Months)
RANO-BM Measurable
RANO-BM Non-measurable

EGFR, epidermal growth factor receptor; RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; RECIST, Response Evaluation Criteria in Solid Tumors.

- Median PFS per RECIST v1.1 was 5.0 (95% CI: 3.4, 7.1) months for Cohort C total and 5.4 (95% CI: 3.4, 7.4) months for the RANO-BM measurable group (**Figure 5**)

Figure 5. Progression-free survival

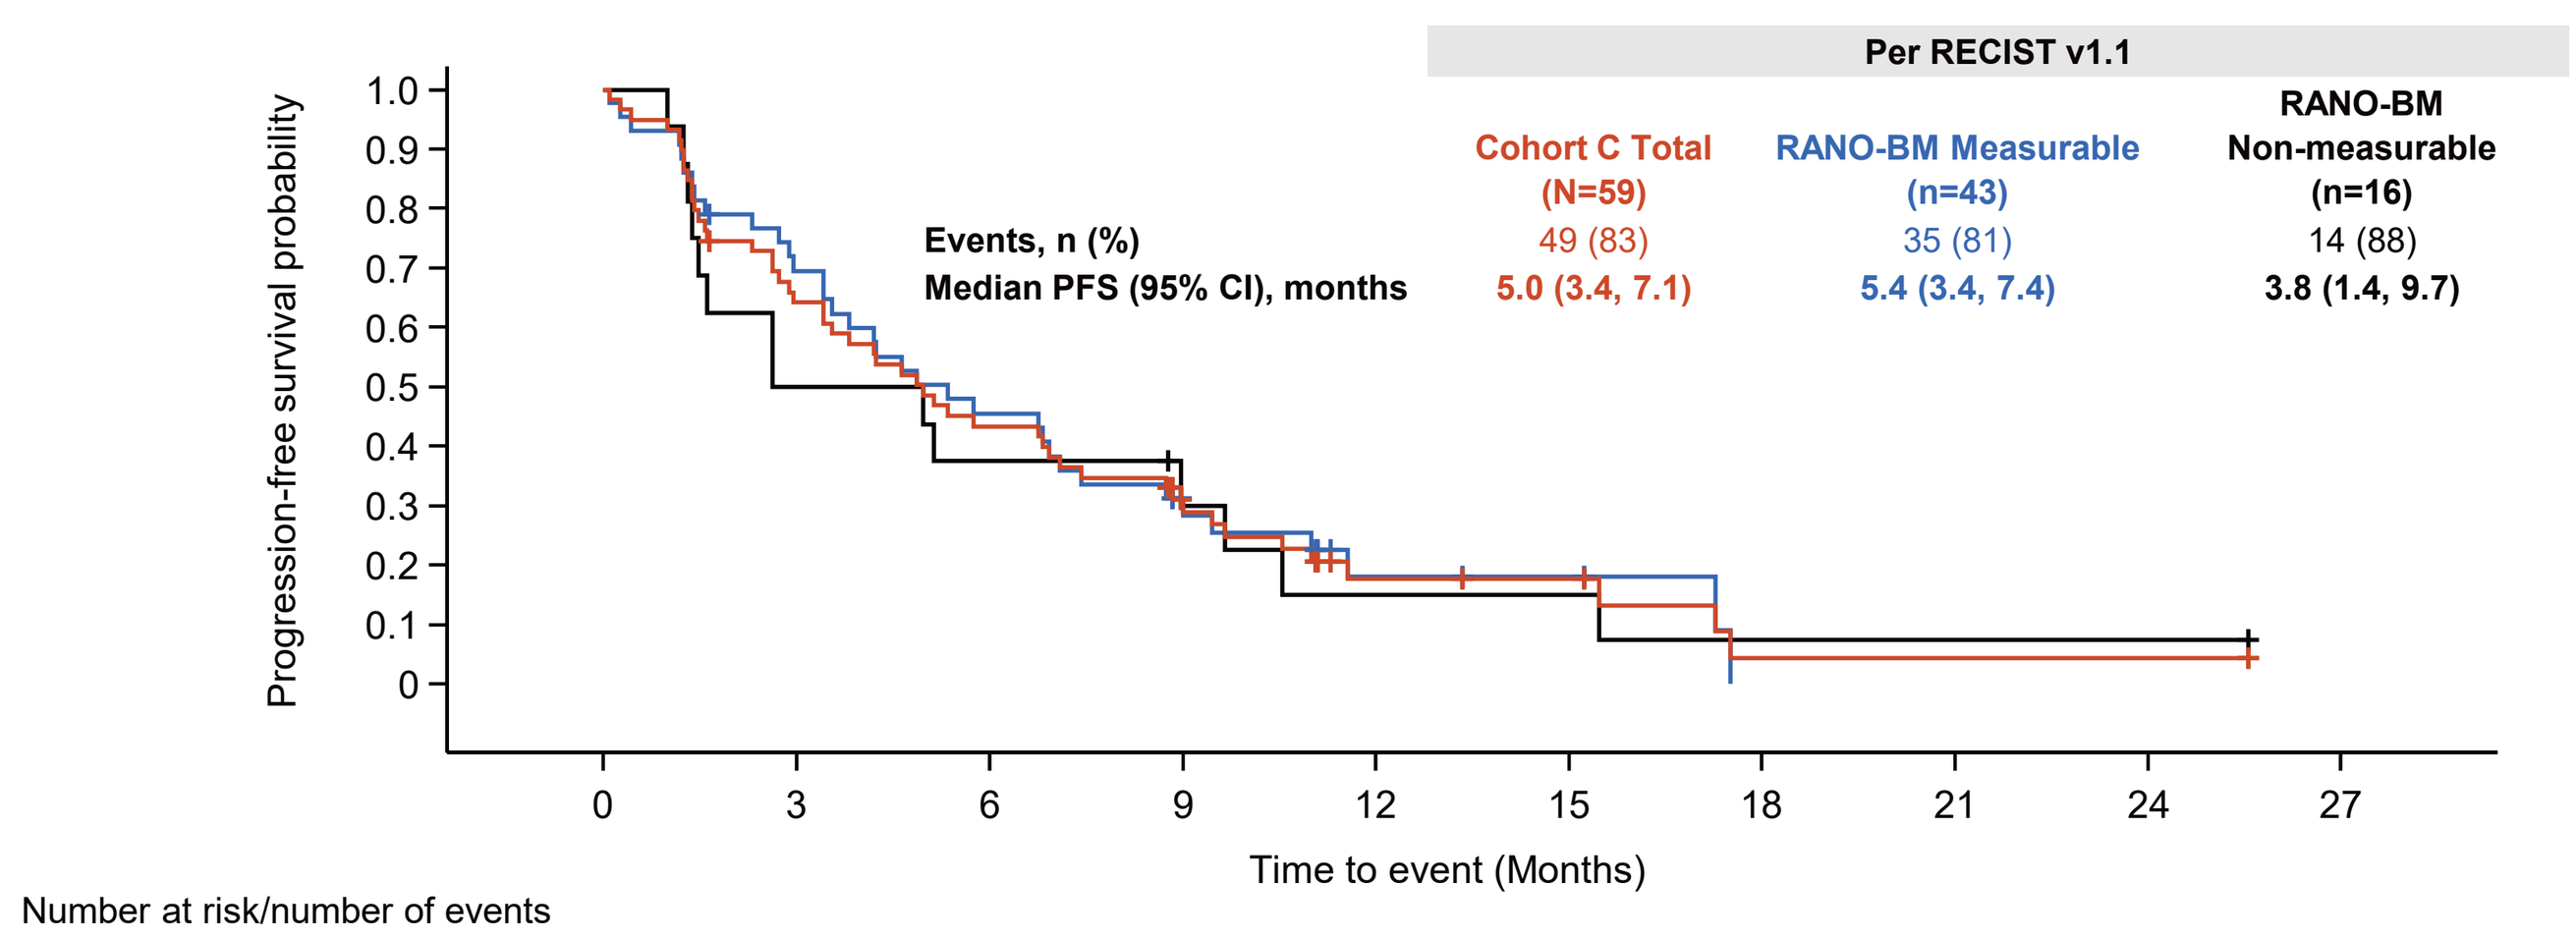

Per RECIST v1.1
Cohort C Total (N=59)
RANO-BM Measurable (n=43)
RANO-BM Non-measurable (n=16)
Events, n (%) 49 (83) 35 (81) 14 (88)
Median PFS (95% CI), months 5.0 (3.4, 7.1) 5.4 (3.4, 7.4) 3.8 (1.4, 9.7)
Progression-free survival probability
Time to event (Months)

Number at risk/number of events	0	3	6	9	12	15	18	21	24	27
Cohort C Total	59/0	37/21	25/12	15/7	6/6	5/0	1/3	1/0	1/0	0/0
RANO-BM Measurable	43/0	29/13	19/10	11/6	4/4	2/0	1/1	1/0	1/0	0/0
RANO-BM Non-measurable	16/0	8/8	6/2	4/1	2/2	2/0	1/1	1/0	1/0	0/0

PFS, progression-free survival; RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; RECIST v1.1, Response Evaluation Criteria in Solid Tumors, Version 1.1.

Results: Safety

- Safety profile of zipalertinib 100 mg BID was consistent with previous studies (**Tables 3** and **4**)
- Low incidence of EGFR-related Grade ≥3 toxicity in both Cohort C total and RANO-BM measurable groups
- Grade ≥3 treatment-related adverse events (TRAEs) (in >1 patient) were dermatitis acneiform (n=3), amylase increase (n=2), and interstitial lung disease (n=2) in Cohort C total and dermatitis acneiform (n=2), and amylase increase (n=2) in the RANO-BM measurable group
- Adverse events were manageable (**Tables 3** and **4**)
- Dose reductions, interruptions, or treatment discontinuations due to TRAEs were reported in 7 (12%), 14 (24%), and 7 (12%) patient(s), respectively, in Cohort C total and 5 (12%), 10 (23%), and 5 (12%) patient(s), respectively, in the RANO-BM measurable group
- Grade ≥3 TRAEs leading to treatment discontinuation were interstitial lung disease (n=2), and pneumonitis, anaphylactic reaction, and nausea (n=1 each) in Cohort C total and pneumonitis, anaphylactic reaction, interstitial lung disease, and nausea (n=1 each) in the RANO-BM measurable group

Table 3. Treatment-related adverse events: Cohort C total (N=59)

Any-grade TRAEs in ≥15% of patients, n (%) (N=59)	Any Grade	Grade 1	Grade 2	Grade ≥3
≥1 TRAE	52 (88)	17 (29)	20 (34)	15 (25)
Paronychia	17 (29)	7 (12)	9 (15)	1 (2)
Dermatitis acneiform	13 (22)	5 (9)	5 (9)	3 (5)
Diarrhea	13 (22)	9 (15)	3 (5)	1 (2)
Stomatitis	12 (20)	10 (17)	2 (3)	0
Dry skin	10 (17)	5 (9)	5 (9)	0
Rash	10 (17)	4 (7)	6 (10)	0
Anemia	9 (15)	6 (10)	0	3 (5)

TRAEs, treatment-related adverse events.

Table 4. Treatment-related adverse events: RANO-BM evaluable (n=43)

Any-grade TRAEs in ≥15% of patients, n (%) (n=43)	Any Grade	Grade 1	Grade 2	Grade ≥3
≥1 TRAE	38 (88)	11 (26)	17 (40)	10 (23)
Paronychia	14 (33)	6 (14)	7 (16)	1 (2)
Diarrhea	11 (26)	7 (16)	3 (7)	1 (2)
Dermatitis acneiform	8 (19)	3 (7)	3 (7)	2 (5)
Rash	7 (16)	3 (7)	4 (9)	0
Stomatitis	7 (16)	7 (16)	0	0

RANO-BM, Response Assessment in Neuro-Oncology Brain Metastases; TRAEs, treatment-related adverse events.

CONCLUSIONS

- Zipalertinib demonstrated clinically meaningful intracranial antitumor activity, with a **30% intracranial ORR** by RANO-BM criteria in patients with NSCLC harboring *EGFR* ex20ins or other uncommon single or compound uncommon mutations
 - Zipalertinib also appears to demonstrate **activity in patients with LMD**
- Zipalertinib showed systemic (RECIST 1.1 ORR 37.5%) and intracranial (RANO-BM ORR 34.5%) **activity in patients with ex20ins NSCLC and brain metastases**
- In ex20ins NSCLC, responses occurred after prior amivantamab-only treatment (RECIST v1.1 4/7; RANO-BM 3/6); data in patients pretreated with ex20ins-targeted TKIs was limited
- Response rates were modest in predominantly EGFR TKI-pretreated non-ex20ins NSCLC; small subgroups limit interpretation
- The safety profile of zipalertinib was consistent with previous reports

Presenter Disclosures

Consulting or Advisory Role: AstraZeneca, Daiichi Sankyo, Blueprint Medicines, Janssen, C4 Therapeutics, Cullinan Oncology, Black Diamond Therapeutics, Taiho Oncology, AbbVie, Novocure, Takeda, Bristol Myers Squibb/Roche, Orion Clinical

Research Funding: AstraZeneca (Inst), Astellas Pharma (Inst), Lilly (Inst), Novartis (Inst), Pfizer (Inst), Daiichi Sankyo (Inst), Cullinan Oncology (Inst), Janssen Oncology (Inst), Erasca, Inc (Inst), Blueprint Medicines (Inst), Black Diamond Therapeutics (Inst), Systimmune(Inst)

Other Relationship: Astellas Pharma

References
1. Malholtra J, et al. *JAMA Oncol*. 2025;11:60–9.
2. Watanabe N, et al. *Ann Transl Med*. 2022;10:1283.
3. Hasako S, et al. *Mol Cancer Ther*. 2018;17:1648–58.
4. Udagawa H, et al. *Mol Cancer Res*. 2019;17:2233–43.
5. Piotrowska Z, et al. *J Clin Oncol*. 2025;43:2387–97.

Acknowledgements
The authors would like to thank and acknowledge all trial participants, their families, and trial personnel for participating in the studies. Medical writing assistance was provided by Yvonne E. Yarker, PhD, FRSM, from Medicite LLC, funded by Taiho Oncology, Inc.

Contact email: YuH@mskcc.org

Study sponsored by Taiho Oncology, Inc.

Presented at the European Society for Medical Oncology (ESMO) 2026 Congress; 23–27 October 2026; Madrid, Spain

Copies of this poster obtained through QR, AR, and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.