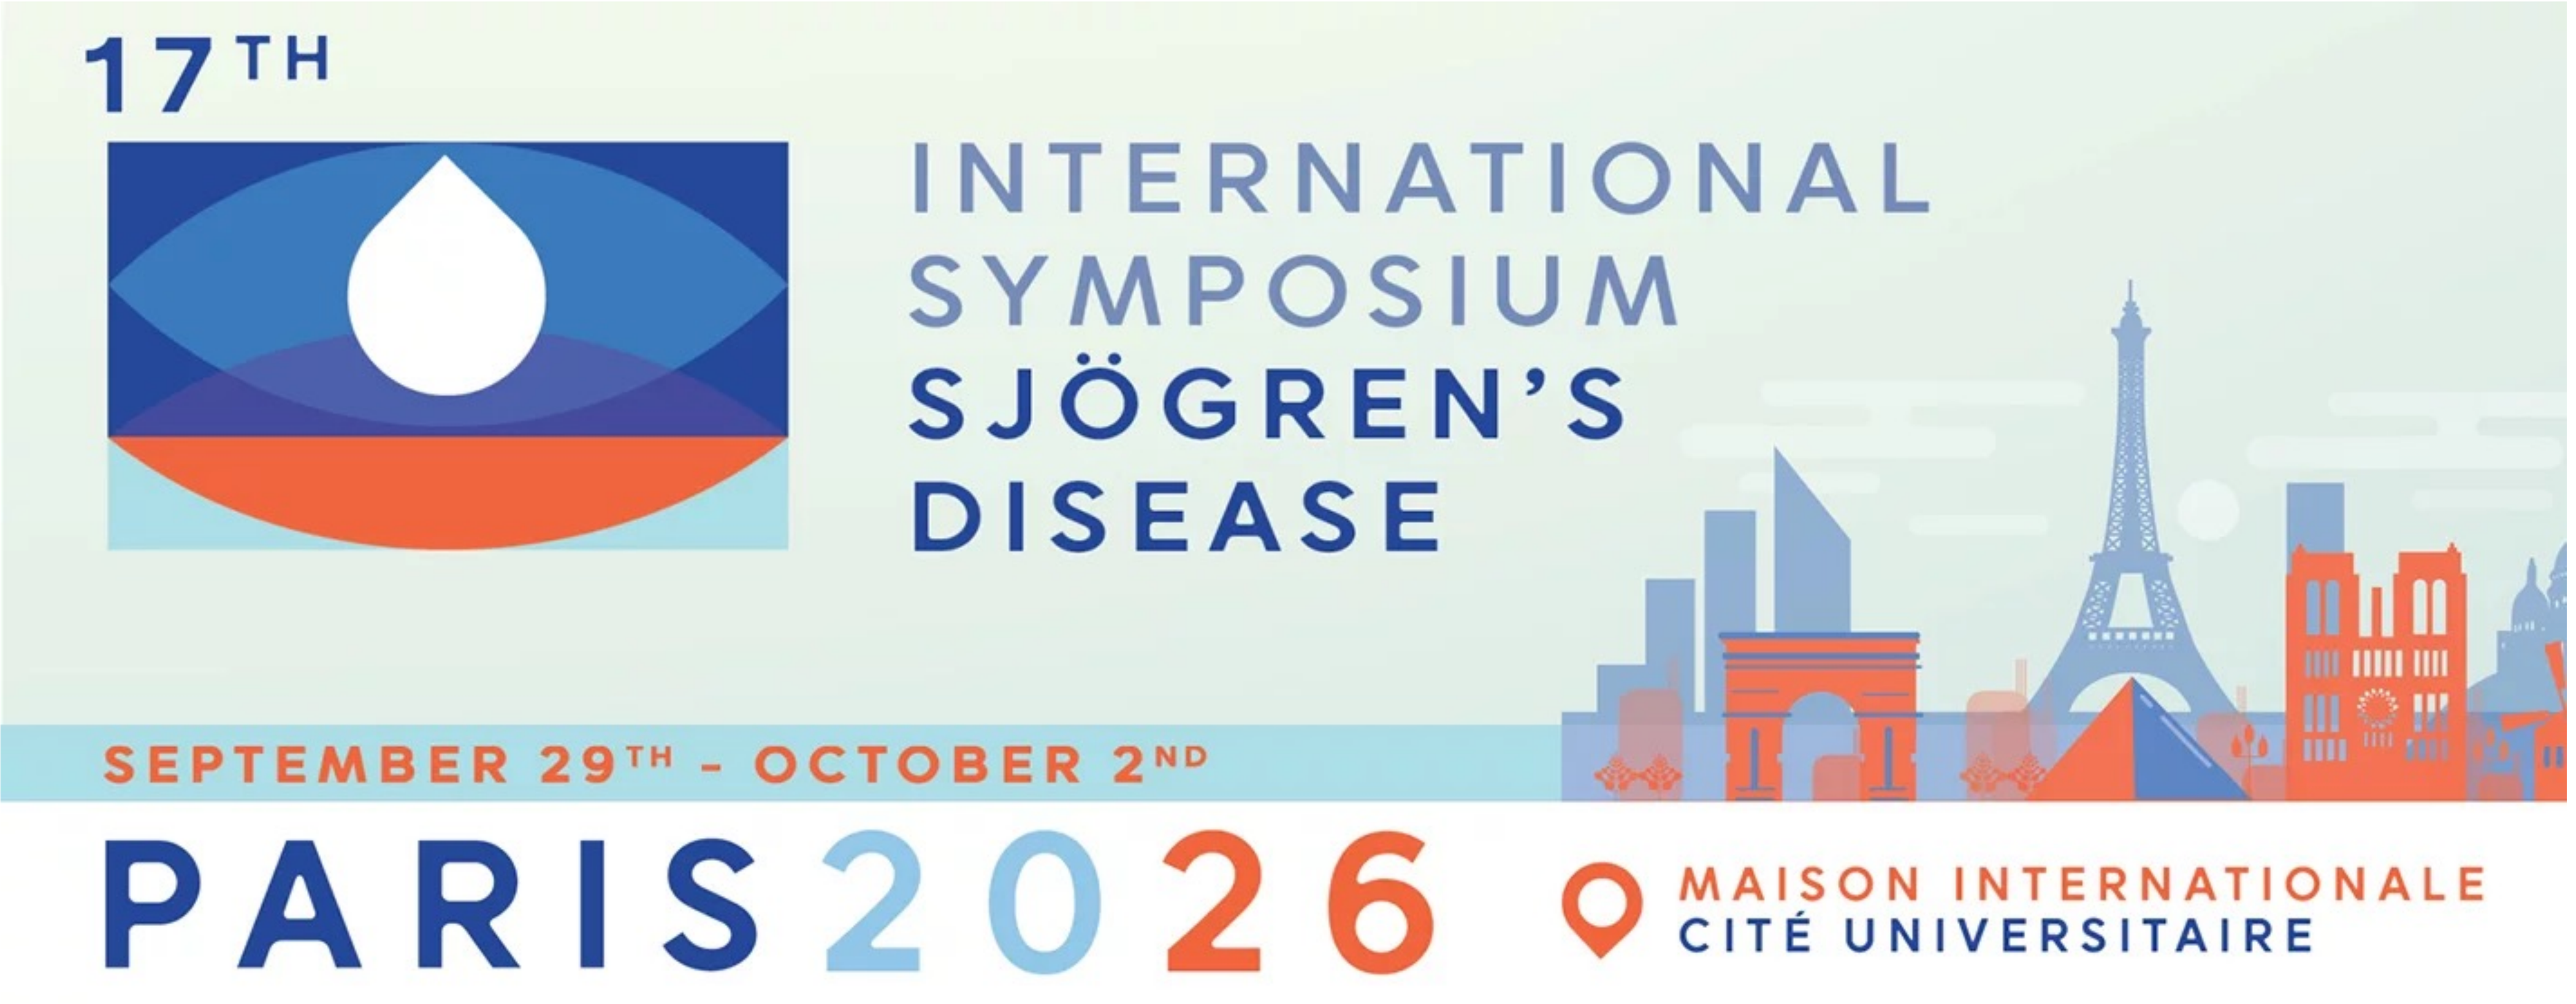
17TH
INTERNATIONAL
SYMPOSIUM
SJÖGREN'S
DISEASE
SEPTEMBER 29TH - OCTOBER 2ND
PARIS 2026
MAISON INTERNATIONALE
CITÉ UNIVERSITAIRE

OUTRACE Sjögren's: A Phase 1b Study of CLN-978, a Bispecific CD19 × CD3 T-Cell Engager, in Patients With Active Sjögren's Disease

Divi Cornec,[1,] Valerie Devauchelle-Pensec,[1] Yue Zhang,[2] Nedal Adi,[3] Kwabena Ayesu,[4] Maria Antonietta D'Agostino,[5] Jacques-Eric Gottenberg,[6,7] Ricardo Grieshaber-Bouyer,[8-10] Pankhuri Gupta,[11] Martin Killian,[12-14] John Lavery,[15] Xavier Mariette,[16,17] Antoine Neel,[18] Debendra Pattanaik,[19] Torsten Witte,[20] Jackie Bronicki,[2] Amy Gubits,[2] Steve Wax,[2] Brian Tozzi,[2] Jeff Jones,[2] Kaida Wu,[2] Antoine Sreih[2]

[1]LBAI, UMR1227, Univ Brest, Inserm, Department of Rheumatology, CHU Brest, Brest, France; [2]Cullinan Therapeutics, Inc, Cambridge, MA, USA; [3]Tranquil Clinical Research, Webster, TX, USA; [4]Omega Research, Orlando, FL, USA; [5]Division of Rheumatology, Fondazione Policlinico Universitario A. Gemelli IRCCS, Università Cattolica del Sacro Cuore, Rome, Italy; [6]Service de Rhumatologie, Hôpital de Hautepierre, Centre National de Référence des Maladies Auto-Immunes et Systémiques Rares (CNR RESO), Strasbourg, France; [7]Laboratoire d'Immunopathologie et de Chimie Thérapeutique, Institut de Biologie Moléculaire et Cellulaire (IBMC), CNRS UPR3572, Strasbourg, France; [8]Department of Internal Medicine 3, Universitätsklinikum Erlangen, Friedrich-Alexander Universität (FAU) Erlangen-Nuremberg, Erlangen, Germany; [9]Deutsches Zentrum Immuntherapie, Friedrich-Alexander Universität Erlangen-Nürnberg (FAU), Erlangen, Germany; [10]Universitätsklinikum Erlangen, Erlangen, Germany; [11]Rheumatology, Intermountain Medical Center, Murray, UT, USA; [12]Department of Internal Medicine, CHU de Saint-Étienne, Saint-Étienne, France; [13]CIRI (Centre International de Recherche en Infectiologie), Team GIMAP, Université Jean Monnet, Université Claude Bernard Lyon 1, Université de Lyon, INSERM U1111, CNRS UMR530, Saint-Étienne, France; [14]Clinical Investigation Center, INSERM CIC-EC 1408, Saint-Etienne, France; [15]Stryde Research, Frisco, TX, USA; [16]Department of Rheumatology, Hôpital Bicêtre, AP-HP, FHU CARE, INSERM UMR 1184, CEA, Université Paris-Saclay, Le Kremlin-Bicêtre, France; [17]Center for Immunology of Viral Infections and Autoimmune Diseases (IMVA), Institut pour la Santé et la Recherche Médicale (INSERM) UMR 1184, Université Paris-Saclay, Paris, France; [18]Internal Medicine Department, Hôtel-Dieu, Nantes University Hospital, Nantes, France; [19]University of Tennessee Health Science Center, Memphis, TN, USA; [20]Klinik für Rheumatologie und Immunologie, Medizinische Hochschule Hannover (MHH), Hannover, Germany

INTRODUCTION

- Targeting CD19-expressing B cells has demonstrated clinical benefit across autoimmune diseases, including Sjögren's disease (SjD)[1–3]
- CLN-978 is a subcutaneously administered CD19×CD3 T-cell engager with a high affinity to CD19 and an albumin-binding domain to extend half-life (**Figure 1**)
- In recent phase 1b clinical trials, a single target dose of CLN-978 induced robust B cell depletion in both peripheral blood and tissues, with promising clinical efficacy in patients with rheumatoid arthritis and systemic lupus erythematosus at the initial dose levels tested[5]
- These data support the evaluation of CLN-978 in patients with SjD

HOW CLN-978 WORKS

- CLN-978 binds CD3 on T cells and CD19 on B cells
- T cell activation induces release of cytotoxic proteins (granzymes and perforin), resulting in depletion of CD19+ B cells

ADVANTAGES OF T-CELL ENGAGERS

- No patient-specific manufacturing → off-the-shelf
- No preconditioning/lymphodepletion
- Subcutaneous dosing → ease of administration
- Dosing flexibility
- Potential for re-dosing to extend remissions
- Outpatient potential

Figure 1. T-cell engagers drive targeted depletion of autoreactive B cells[4]

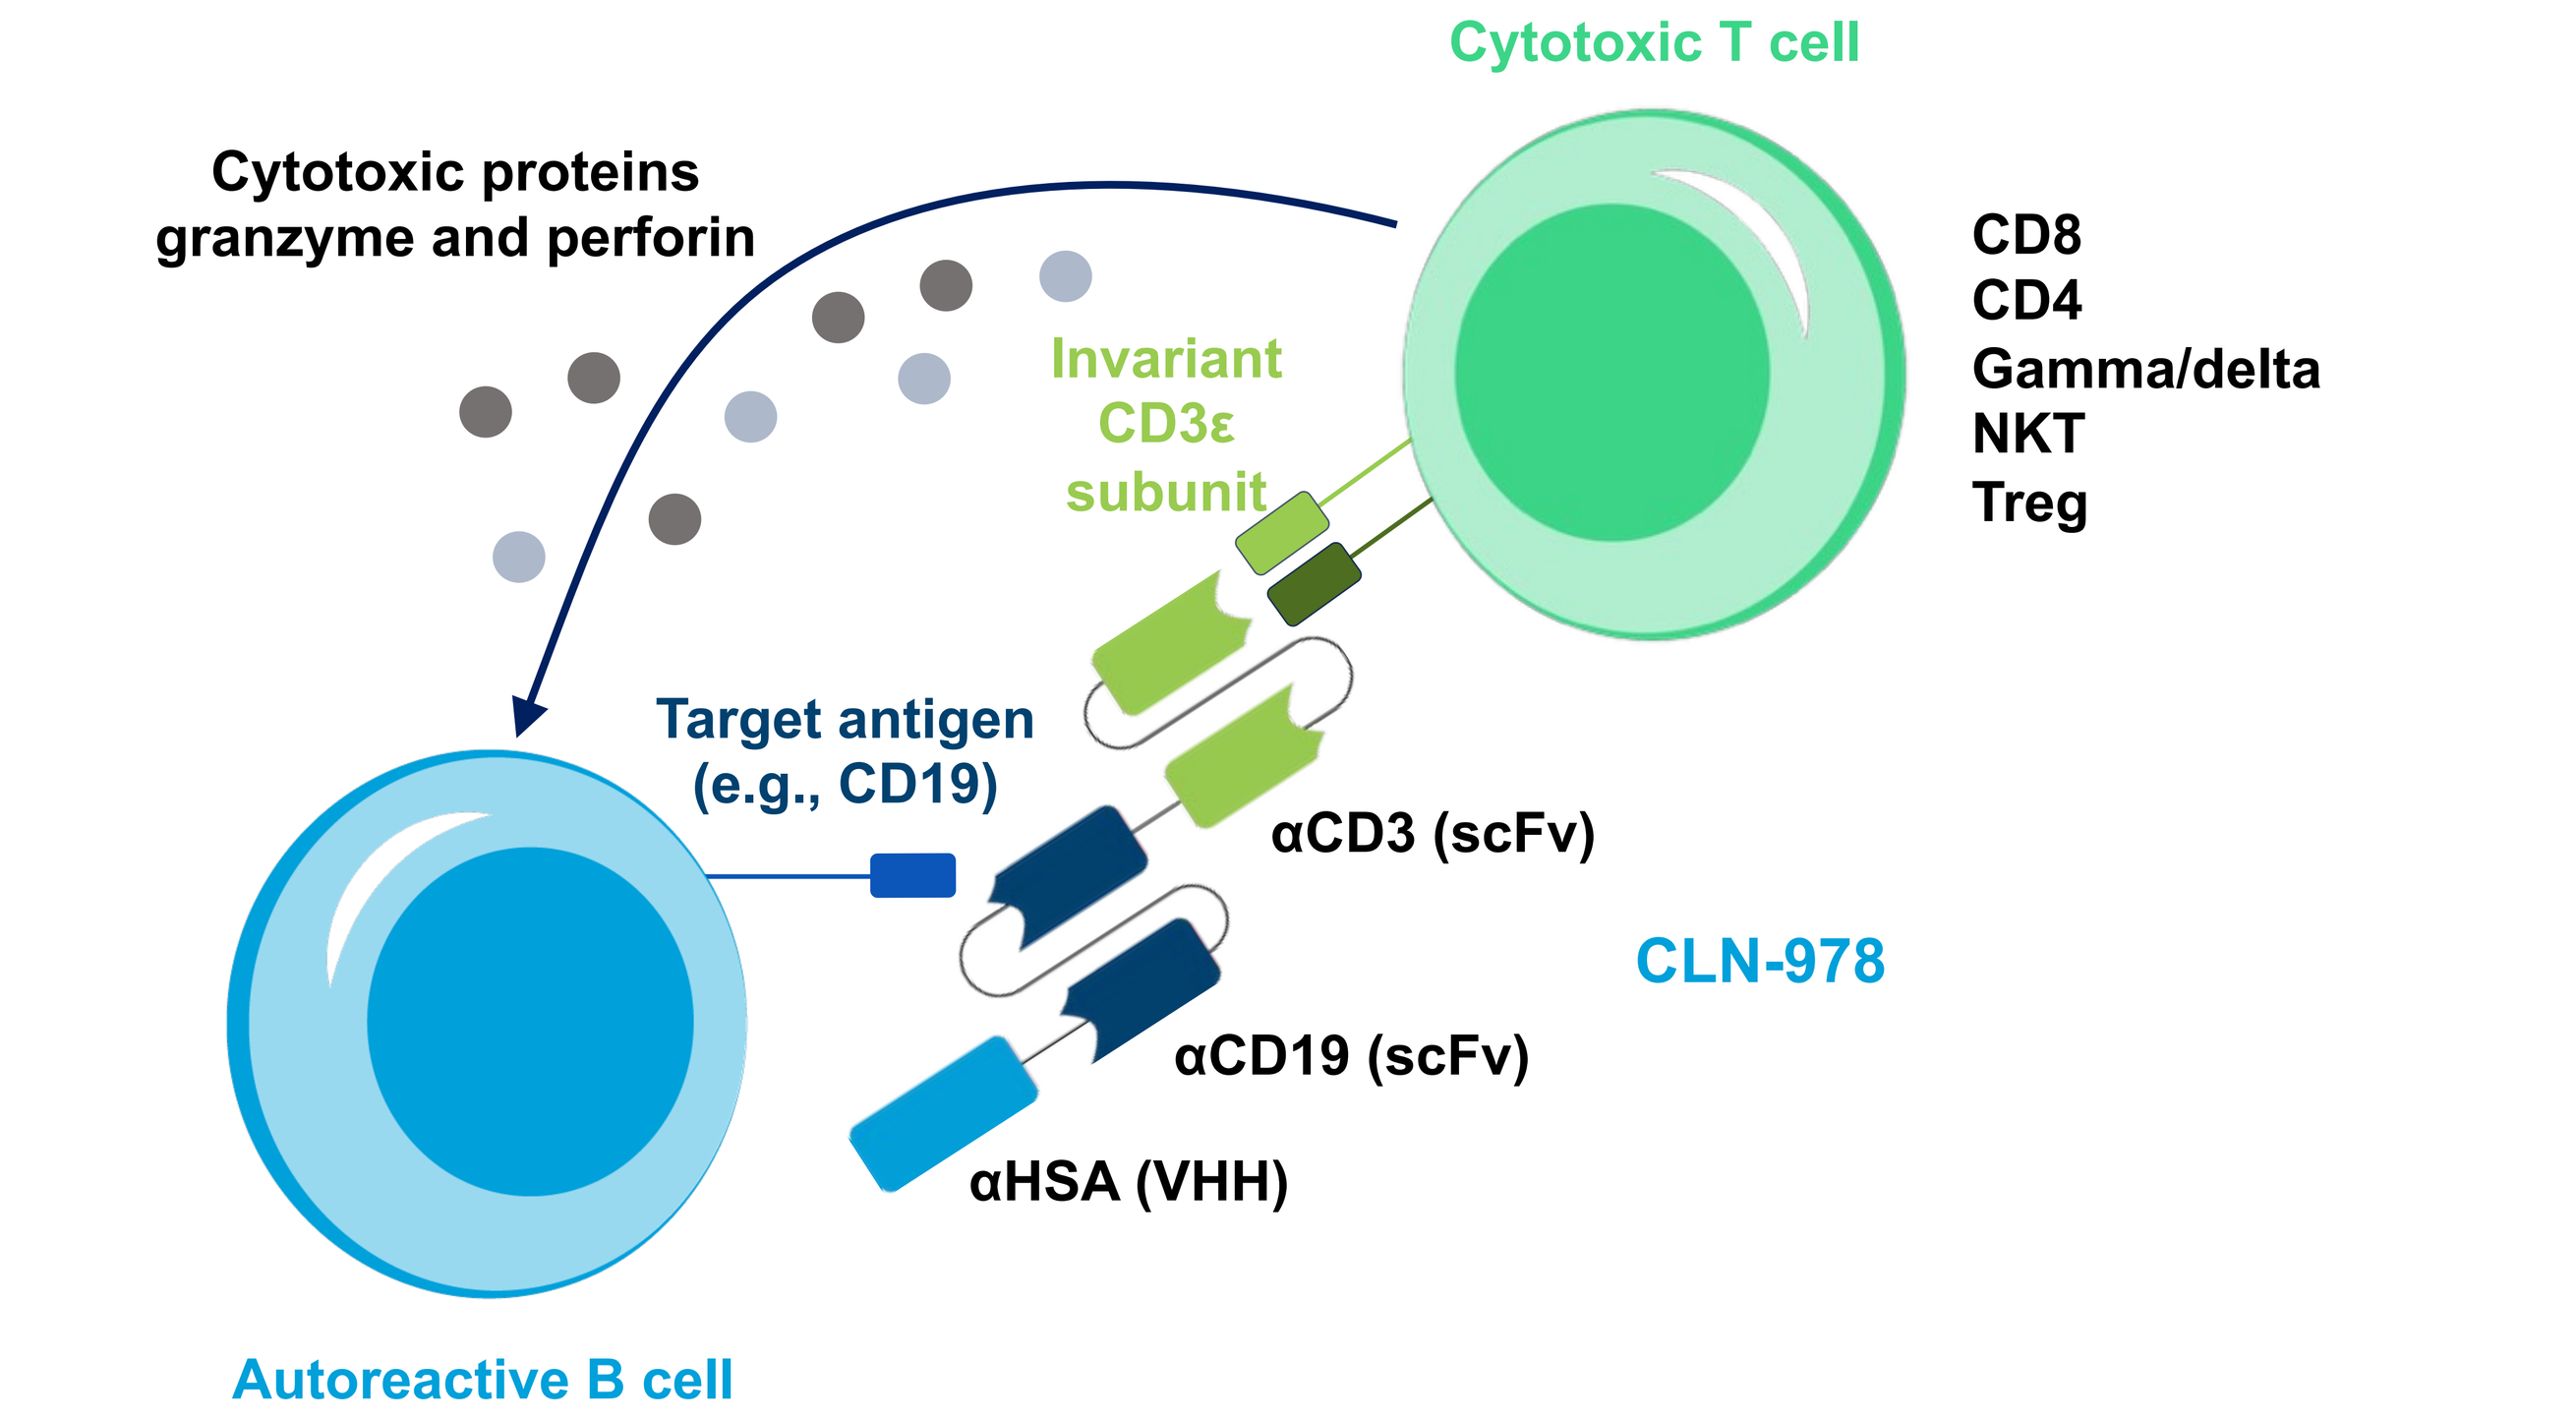
Cytotoxic T cell
Cytotoxic proteins
granzyme and perforin
Invariant
CD3ε
subunit
CD8
CD4
Gamma/delta
NKT
Treg
Target antigen
(e.g., CD19)
αCD3 (scFv)
CLN-978
αCD19 (scFv)
αHSA (VHH)
Autoreactive B cell

HSA, human serum albumin; NKT, natural killer T cell; scFv, single-chain variable fragment; Treg, regulatory T cell; VHH, variable heavy domain.

METHODS

- OUTRACE Sjögren's (NCT07041099; EUCTR 2025-521462-96-00) is an ongoing, Phase 1b study designed to evaluate the safety, pharmacokinetics (PK), pharmacodynamics (PD), immunogenicity, and preliminary clinical activity of CLN-978 in patients with active SjD
- The study consists of 2 parts (**Figure 2**)
 - Part A dose escalation evaluates step-up dosing, beginning with a priming dose followed by escalating weekly doses to identify a recommended dose and regimen
 - Part B dose expansion will further evaluate safety, PK, PD, and clinical activity at selected dose levels from Part A and will explore alternative dosing schedules
- CLN-978 is administered during a 4-week treatment phase, followed by longitudinal assessments through Week 48, with optional long-term follow-up

Figure 2. OUTRACE Sjögren's (CLN-978-SJ-101) is a Phase 1b, multicenter, open-label, dose-escalation and dose-expansion study

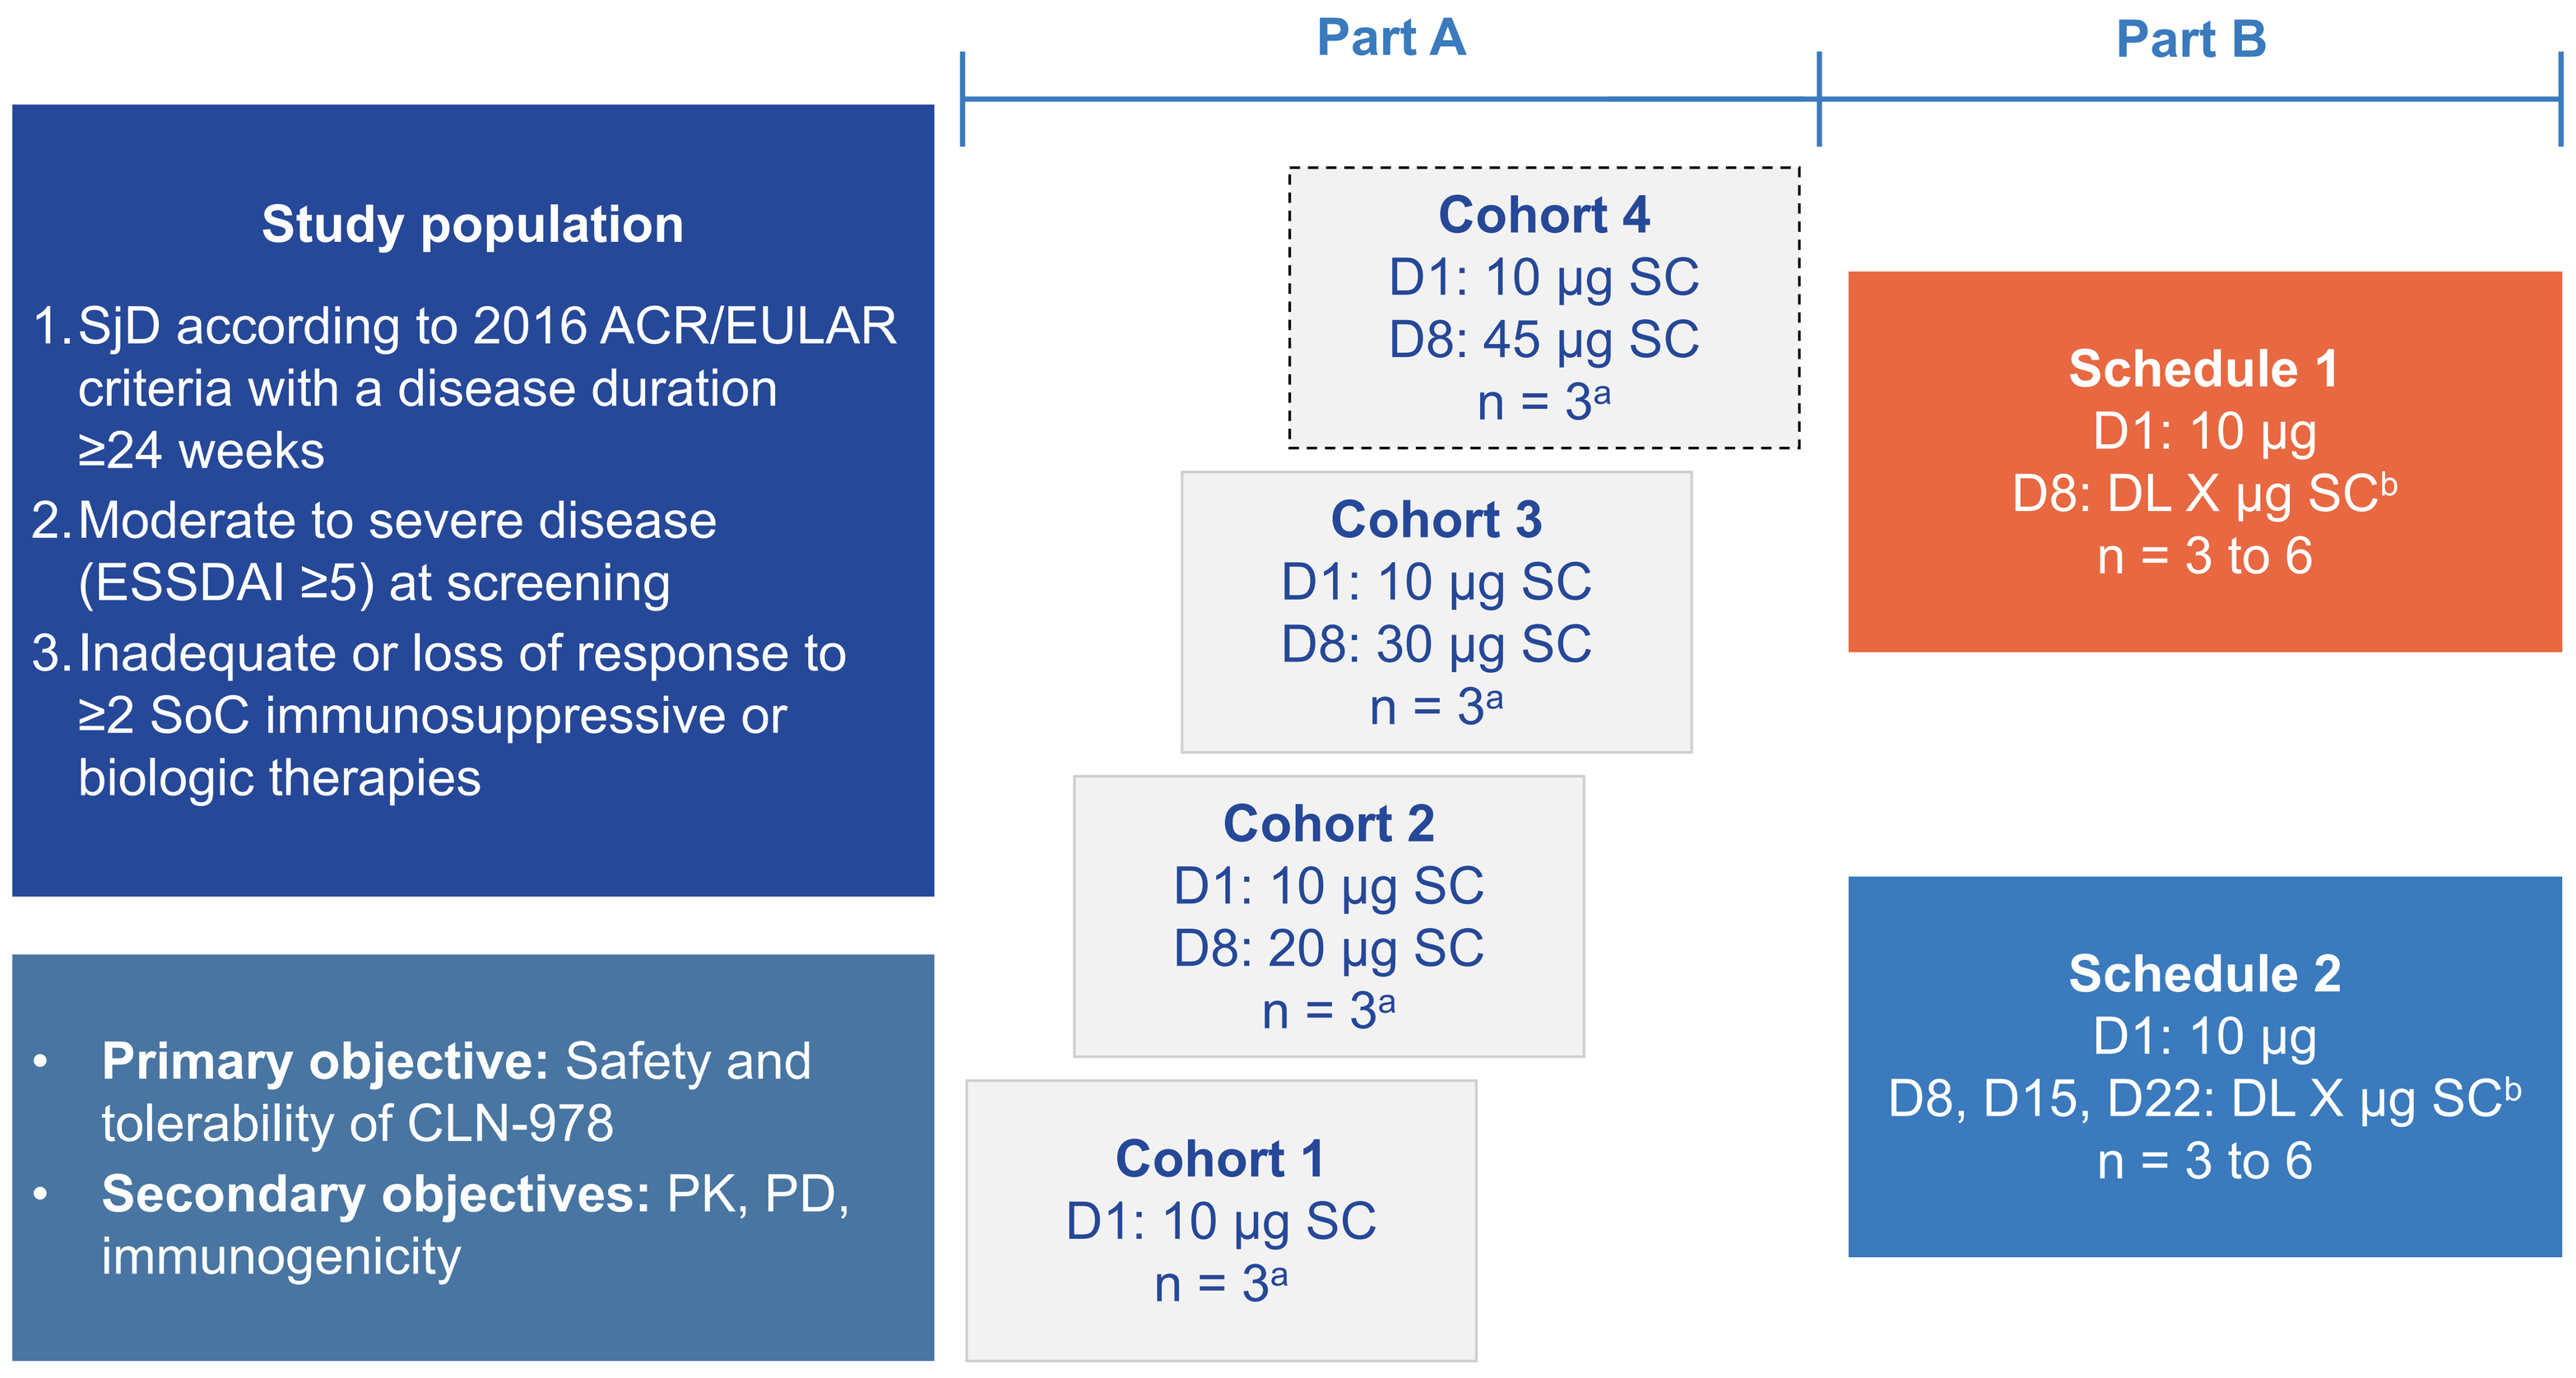
Part A
Part B
Study population
1. SjD according to 2016 ACR/EULAR criteria with a disease duration ≥24 weeks
2. Moderate to severe disease (ESSDAI ≥5) at screening
3. Inadequate or loss of response to ≥2 SoC immunosuppressive or biologic therapies
Primary objective: Safety and tolerability of CLN-978
Secondary objectives: PK, PD, immunogenicity
Cohort 4
D1: 10 µg SC
D8: 45 µg SC
n = 3a
Cohort 3
D1: 10 µg SC
D8: 30 µg SC
n = 3a
Cohort 2
D1: 10 µg SC
D8: 20 µg SC
n = 3a
Cohort 1
D1: 10 µg SC
n = 3a
Schedule 1
D1: 10 µg
D8: DL X µg SCb
n = 3 to 6
Schedule 2
D1: 10 µg
D8, D15, D22: DL X µg SCb
n = 3 to 6

ACR, American College of Rheumatology; D, day; DL, dose level; ESSDAI, EULAR Sjögren's Syndrome Disease Activity Index; EULAR, European Alliance of Associations for Rheumatology; PD, pharmacodynamics; PK, pharmacokinetics; SC, subcutaneous; SjD, Sjögren's disease; SoC, standard of care. [a]Up to 3 additional patients may be added following cohort review by the Safety Review Committee. [b]Dose level to be determined based on Part A findings.

Inclusion

- Meets 2016 ACR/EULAR classification criteria for SjD
- Disease duration of ≥24 weeks prior
- Moderate-to-severe disease (ESSDAI ≥5)
- Adequate hematologic, hepatic, and renal function
- Inadequate response or intolerance to ≥2 prior systemic therapies, including immunosuppressants and/or biologics
- Patients with a negative anti-SSA are eligible if the salivary gland biopsy is consistent with SjD

Exclusion

- Concomitant rheumatic disease
- Active or recurrent infection or immunodeficiency
- Severe neurologic disease
- High risk for thrombosis
- Rapidly progressive glomerulonephritis or nephrotic range proteinuria
- History of splenectomy or recent live/attenuated vaccination

	Study objective	Endpoint(s)
Primary	To evaluate safety and tolerability of CLN-978	Incidence and severity of AEs, serious AEs, and AEs of special interest
Secondary	To evaluate the PK of CLN-978	Serum concentrations of CLN-978
	To evaluate the PD of CLN-978	Change from baseline in B lymphocyte cell numbers in peripheral blood
	To evaluate the immunogenicity of CLN-978	Detection of anti-drug antibodies
Exploratory	To evaluate CLN-978 biomarkers	PD markers of B cell depletion
	To evaluate clinical efficacy of CLN-978	ESSDAI, ESSPRI, STAR, CRESS

AE, adverse event; CRESS, Composite of Relevant Endpoints for Sjögren's Syndrome; ESSDAI, EULAR Sjögren's Syndrome Disease Activity Index; ESSPRI, EULAR Sjögren's Syndrome Patient Reported Index; EULAR, European Alliance of Associations for Rheumatology; PD, pharmacodynamics; PK, pharmacokinetics; STAR, Sjögren's Tool for Assessing Response.

Figure 3. The study is currently enrolling patients at sites in the United States, Europe, and Australia

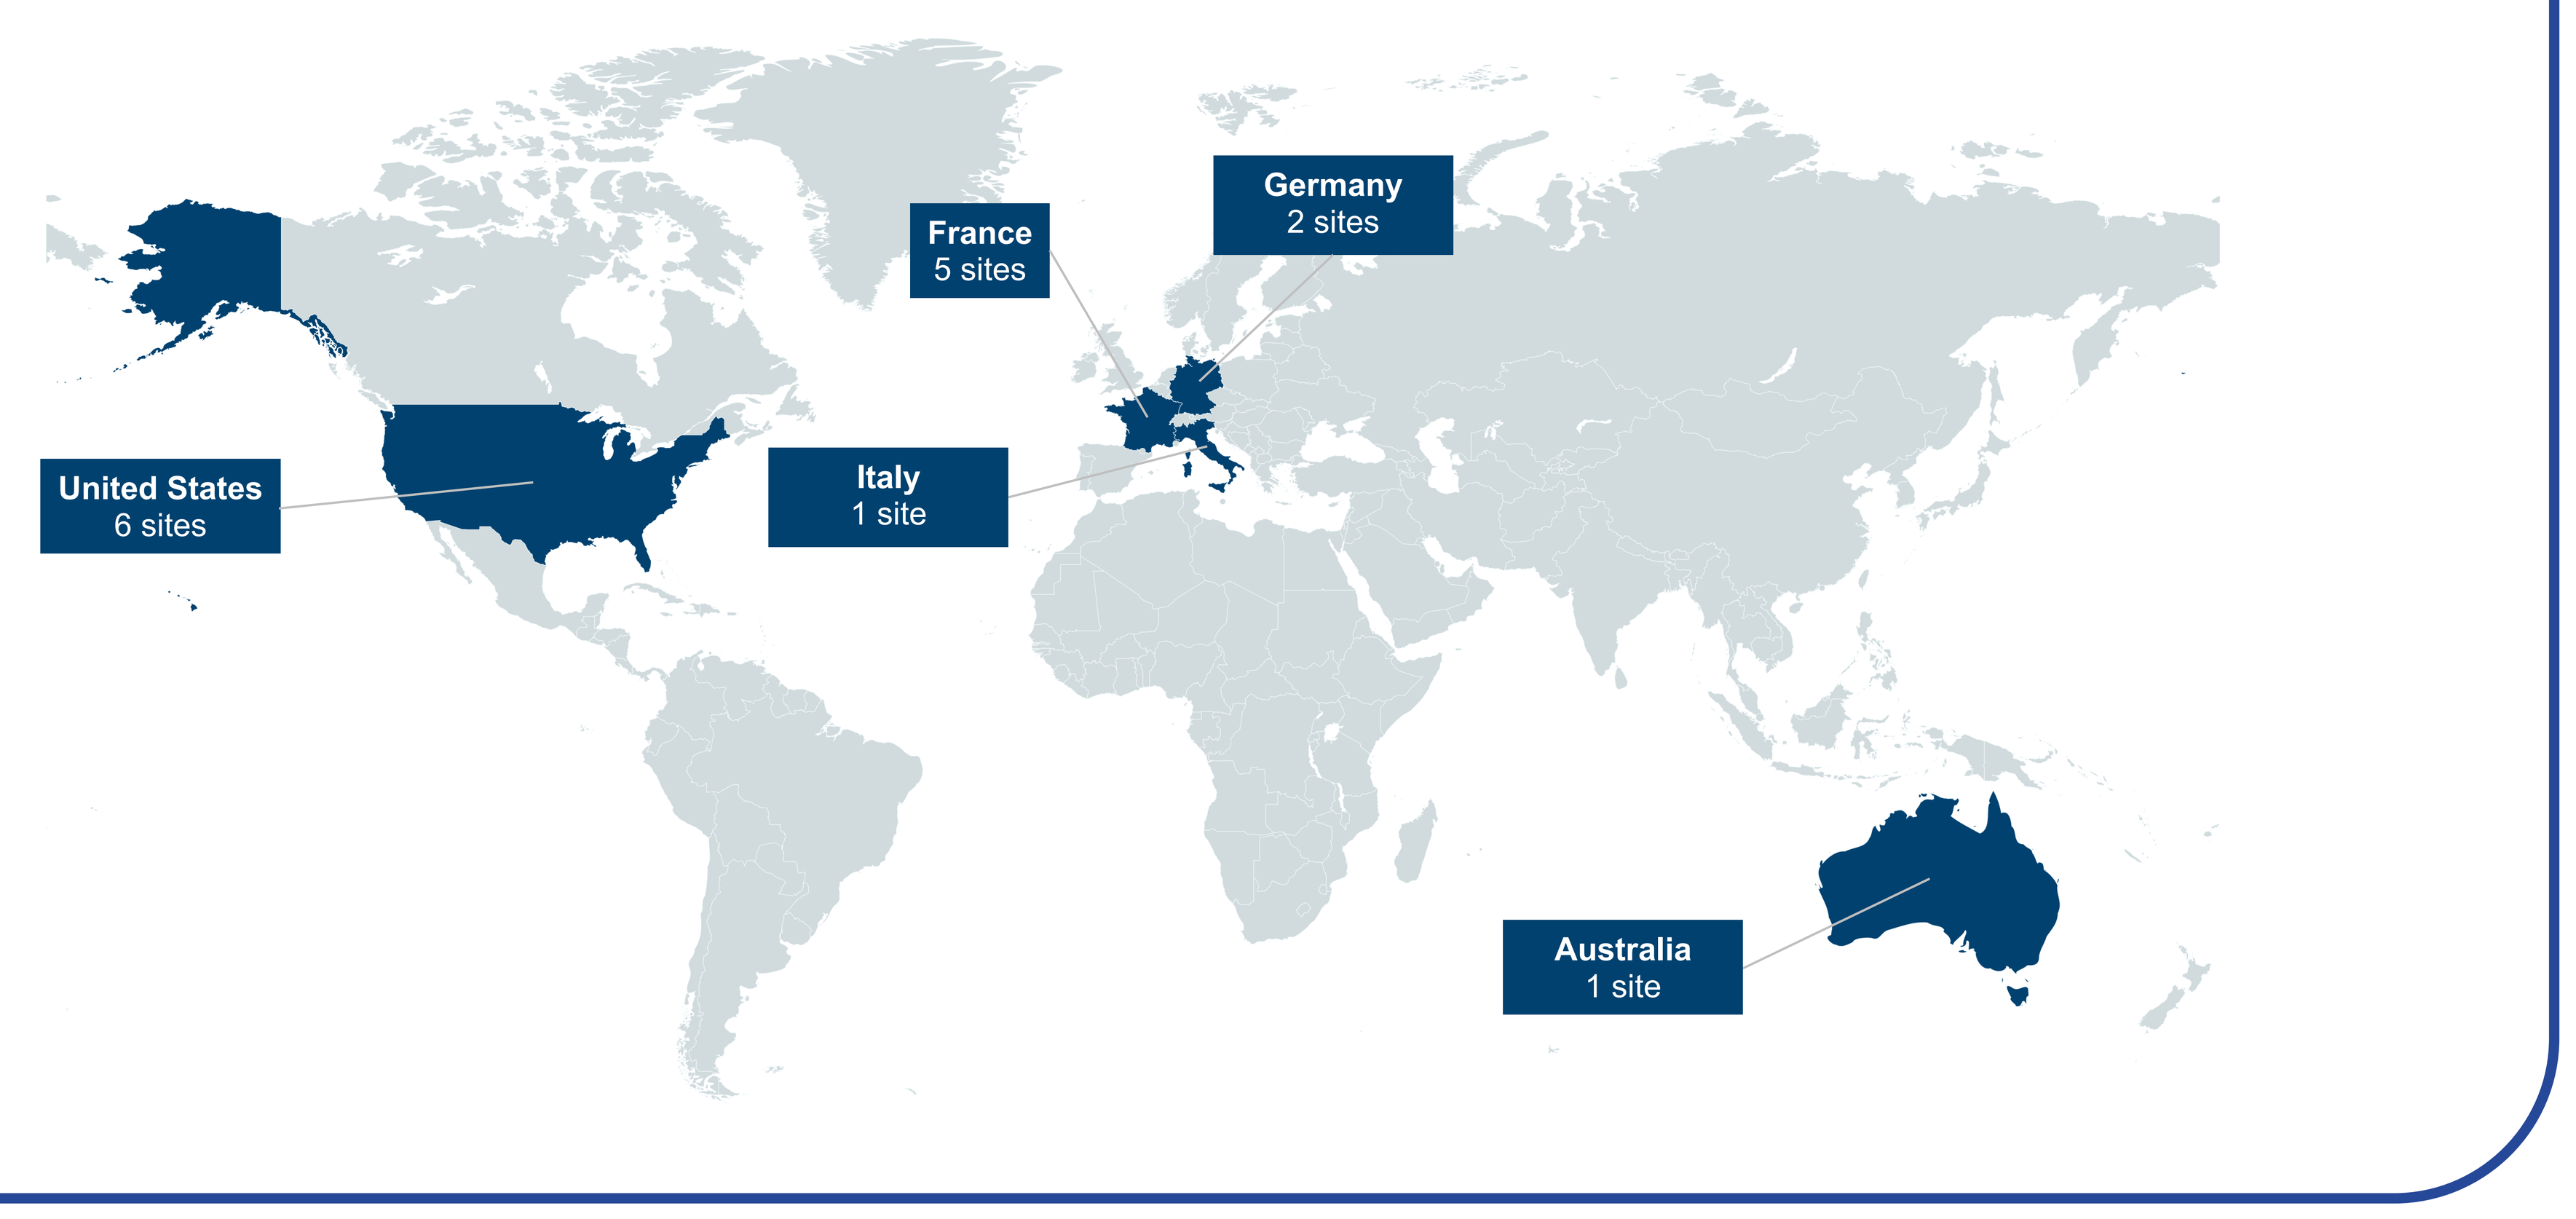
France
5 sites
Germany
2 sites
United States
6 sites
Italy
1 site
Australia
1 site

CONCLUSIONS

- OUTRACE Sjögren's will characterize the safety, PK, and PD of CLN-978 and explore its potential to induce deep peripheral and tissue B cell depletion and improve disease activity in patients with active SjD
- This study will inform dose selection and direct further clinical development of CLN-978 in patients with SjD

REFERENCES

1. Müller F, et al. *N Engl J Med.* 2024;390(8):687–700.
2. Sheng L, et al. *Front Immunol.* 2023;14:1298815.
3. Li Y, et al. *Cell Res.* 2025;35(3):220–223.
4. Nagorsen D, Baeuerle PA. *Exp Cell Res.* 2011;317(9):1255–1260.
5. Grieshaber-Bouyer R, et al. CLN-978, a bispecific CD19 × CD3 T-cell engager, induces robust B-cell depletion in patients with rheumatoid arthritis and systemic lupus erythematosus. Poster presented at: EULAR 2026; June 3–6, 2026; London, United Kingdom.

ACKNOWLEDGEMENT

The authors express sincere gratitude to all participants of this trial. This study is sponsored by Cullinan Therapeutics, Inc. Professional medical writing support was provided by Peloton Advantage, LLC, an OPEN Health company.

CONTACT INFORMATION

Antoine Sreih; asreih@cullinantx.com