CLN-978, a bispecific CD19 x CD3 T-cell engager, induces robust B-cell depletion in patients with rheumatoid arthritis and systemic lupus erythematosus

Ricardo Grieshaber-Bouyer,[1,2,3] Anca Dinu Askanase,[4] Rob Will,[5] Nedal Adi,[6] Kwabena Ayesu,[7] Adrian Reber,[8] Yue Zhang,[8] Sarai Faison,[8] Todd Shearer,[8] Irina Shapiro,[8] Stephen Wax,[8] Jeff Jones,[8] Maria Gabriella Raimondo,[1,2,3] Carlo Tur,[1,2,3,9] Stefano Alivernini,[9] Georg Schett,[1,2,3] Maria Antonietta D'Agostino[9]

[1]Friedrich-Alexander University (FAU) of Erlangen–Nuremberg, Erlangen–Nuremberg, Germany; [2]Universitätsklinikum Erlangen, Department of Internal Medicine, Erlangen, Germany; [3]Deutsches Zentrum Immuntherapie, Universitätsklinikum Erlangen, Erlangen, Germany; [4]Lupus Center at Columbia University Medical Center, Division of Rheumatology, New York, New York, USA; [5]Osteoporosis Solutions, Victoria Park, Western Australia, Australia; [6]Tranquil Clinical Research, Webster, Texas, USA; [7]Omega Research Group, Orlando, Florida, USA; [8]Cullinan Therapeutics, Inc., Cambridge, Massachusetts, USA; [9]Fondazione Policlinico Universitario Agostino Gemelli IRCCS - Università Cattolica del Sacro Cuore, Division of Rheumatology, Rome, Italy

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

POS1179

BACKGROUND

- Systemic lupus erythematosus (SLE) and rheumatoid arthritis (RA) are systemic autoimmune diseases responsive to B-cell–directed treatments[1,2]
- T-cell engagers can induce deep B-cell depletion and offer advantages with off-the-shelf availability and flexible dosing, as compared with cell-based therapies such as CAR T cells[3,4]
- CLN-978 is a subcutaneously administered CD19 x CD3 T-cell engager

T-cell engagers drive targeted depletion of autoreactive B cells[5]

HOW T-CELL ENGAGERS WORK

- Bispecific T-cell engagers bind CD3 on T cells and CD19 on B cells
- T-cell activation induces release of cytotoxic proteins, resulting in depletion of CD19+ B cells

ADVANTAGES OF T-CELL ENGAGERS

- Outpatient potential
- Off-the-shelf, no manufacturing required
- Subcutaneous dosing, ease of administration
- Dosing flexibility
- Potential for re-dosing to extend remissions

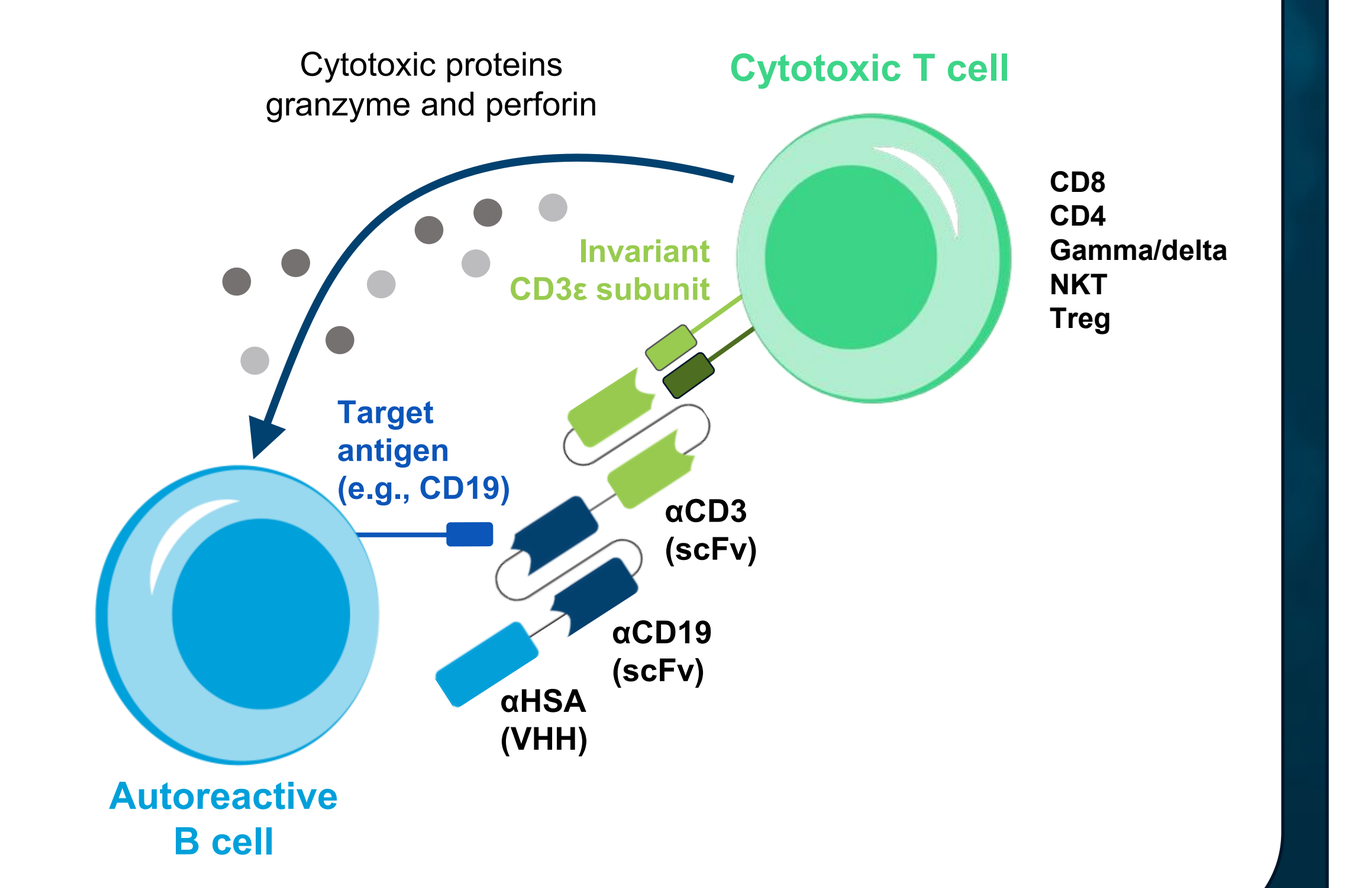

Cytotoxic proteins granzyme and perforin
Cytotoxic T cell
CD8
CD4
Gamma/delta
NKT
Treg
Invariant CD3ε subunit
Target antigen (e.g., CD19)
αCD3 (scFv)
αCD19 (scFv)
αHSA (VHH)
Autoreactive B cell

OBJECTIVE

Evaluate the safety, pharmacodynamics, and effects on disease activity of CLN-978 in two phase 1b, open-label, dose-escalation studies of patients with treatment-refractory SLE (NCT06613360) and RA (NCT06994143)

STUDY DESIGNS

SLE patient population

- SLE meeting 2019 EULAR/ACR criteria
- ≥ 1 of the following SLE auto-antibodies: ANA (≥ 1:80), anti-dsDNA, anti-Smith
- hSLEDAI ≥ 6 at screening
- Inadequate response to ≥ 2 of the following treatments: oral corticosteroids, antimalarials, conventional immunosuppressants, or biologics

RA patient population

- RA meeting 2010 ACR/EULAR criteria
- Seropositive or B-cell infiltrate in synovial biopsy
- DAS28-ESR ≥ 3.2 and ≥ 1 swollen joint
- Inadequate response to ≥ 2 targeted DMARDs (tsDMARD and/or biologic)

Cohort 4
D1: 10 µg SC
D8: 45 µg SC

Cohort 3
D1: 10 µg SC
D8: 30 µg SC

Cohort 2
D1: 10 µg SC
D8: 20 µg SC

Cohort 1
D1: 10 µg SC

- All patients received premedication with corticosteroid, H1 and H2 blockade, and paracetamol
- Treatment regimens with multiple target dose administrations are explored following single target dose escalation (above)

D=day; SC=subcutaneous

PATIENT CHARACTERISTICS

Patient Characteristics	SLE (N = 18)	RA (N = 11)
Sex, n (%)		
Male	0	1 (9.1)
Female	18 (100)	10 (90.9)
Median age (range), years	42 (22-61)	59 (45-72)
Ethnicity, n (%)		
Hispanic or Latino	3 (16.7)	0
Race, n (%)		
American Indian or Alaska Native	1 (5.6)	0
Asian	1 (5.6)	0
Black or African American	4 (22.2)	0
White	9 (50.0)	10 (90.9)
Other/Not reported	3 (16.7)	1 (9.1)
Prior treatment		
Median number of prior immunosuppressants/biologics (range)	3 (2-6)	6 (1-12)
Prior treatment with immunosuppressants, n (%)	17 (94.4)	11 (100)
Prior treatment with biologics (non–B-cell-directed), n (%)	3 (16.7) [a]	11 (100) [b]
Prior treatment with rituximab, n (%)	1 (5.6)	3 (27.2)
Prior treatment with belimumab, n (%)	5 (27.8)	0

[a] Anifrolumab
[b] Non–B-cell-directed biologics include TNF inhibitors, IL-6 inhibitors, T-cell modulator (abatacept), and IL-23 inhibitors.

B-CELL DEPLETION AND REDUCTION OF DISEASE ACTIVITY WITH EARLY DOSE COHORTS

Systemic Lupus Erythematosus

- Peripheral B-cell counts were reduced by > 80% in 14/17 SLE patients (82%) treated in single target dose cohorts (Cohorts 1-4) (**A**) with dose-dependent recovery **(B)**
 - 7 of 14 patients (50%) treated at target doses ≥ 20 µg (Cohorts 2, 3, and 4) achieved peripheral B-cell depletion below the limit of quantification (BLOQ)
- Among 14 patients with ≥ 4 weeks follow-up, a ≥ 4-point reduction in SLEDAI was observed in 10 patients (71%), with 5 achieving a DORIS remission **(C)**
- All markers of disease activity (anti-dsDNA antibody, UPCR, C3, and C4) improved **(D)**

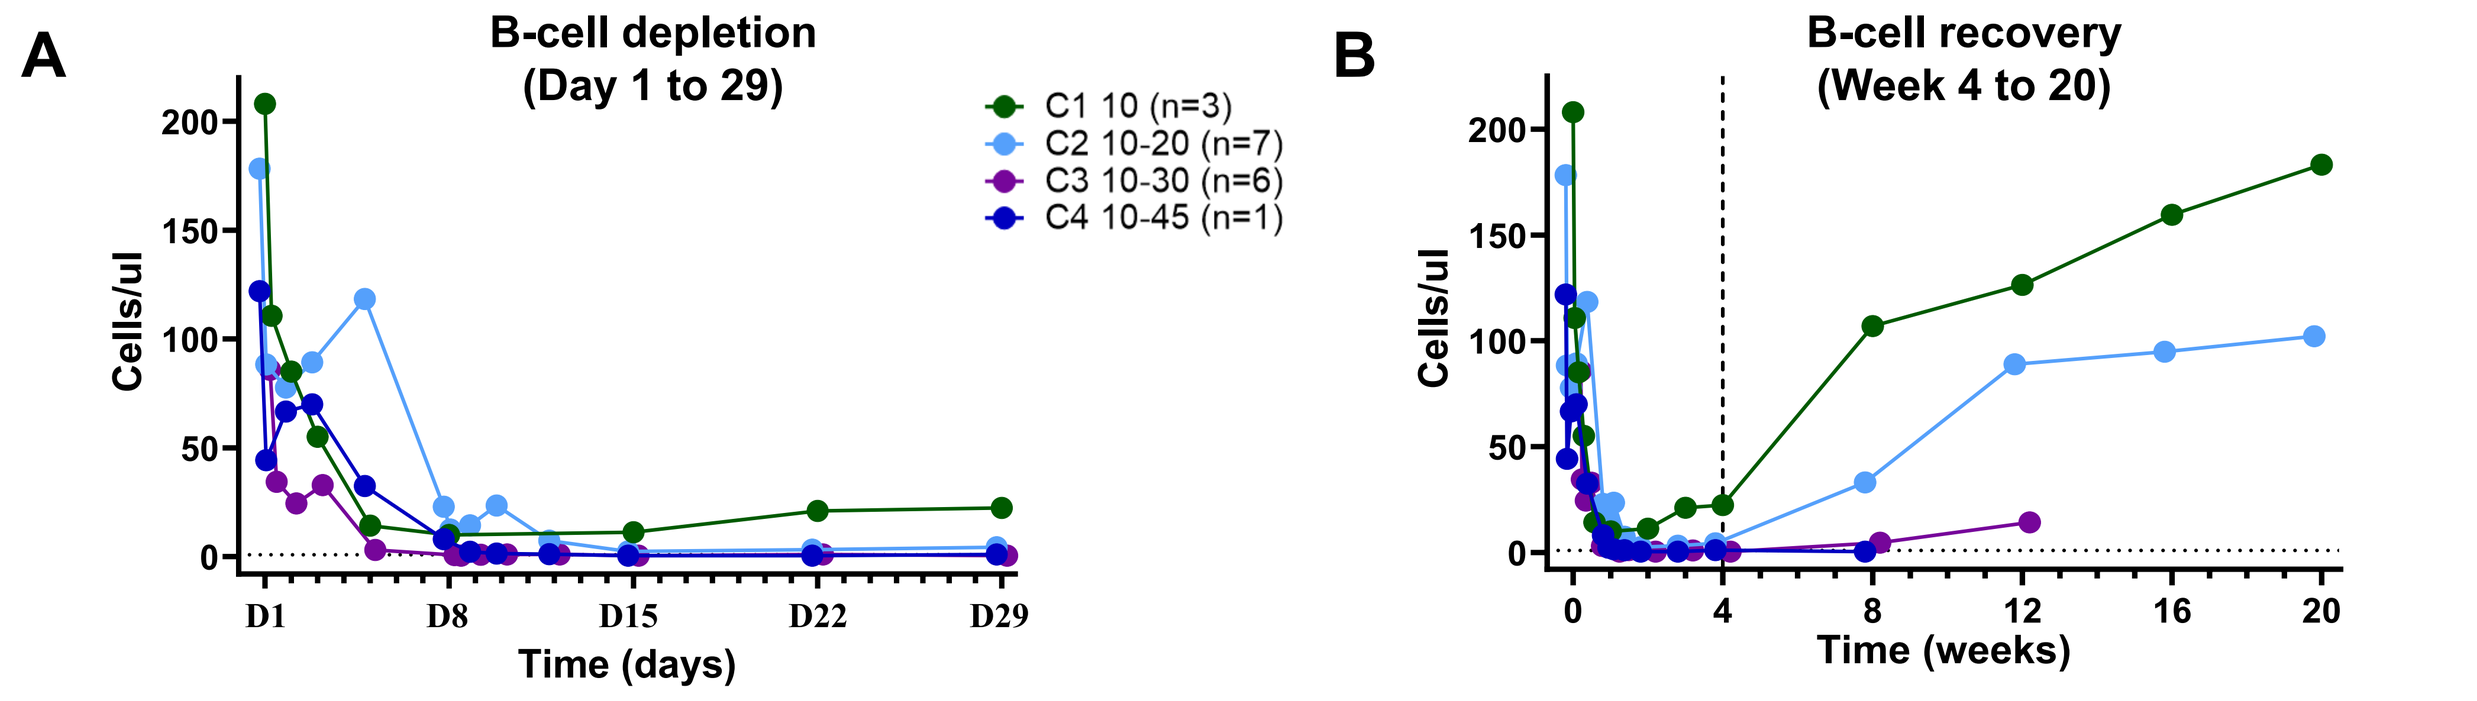

A
B-cell depletion (Day 1 to 29)
C1 10 (n=3)
C2 10-20 (n=7)
C3 10-30 (n=6)
C4 10-45 (n=1)
Cells/ul
Time (days)
B
B-cell recovery (Week 4 to 20)
Time (weeks)

Reduction in B-cell numbers during Days 1-29 after start of treatment (A) and dose-dependent B-cell recovery during weeks 4-20 (B), shown as median per dose level as indicated. Graphs represent patients with available B-cell data through Day 10. B-cell kinetics was measured by a validated TBNK assay with LLOQ of 0.9 cells/µL.

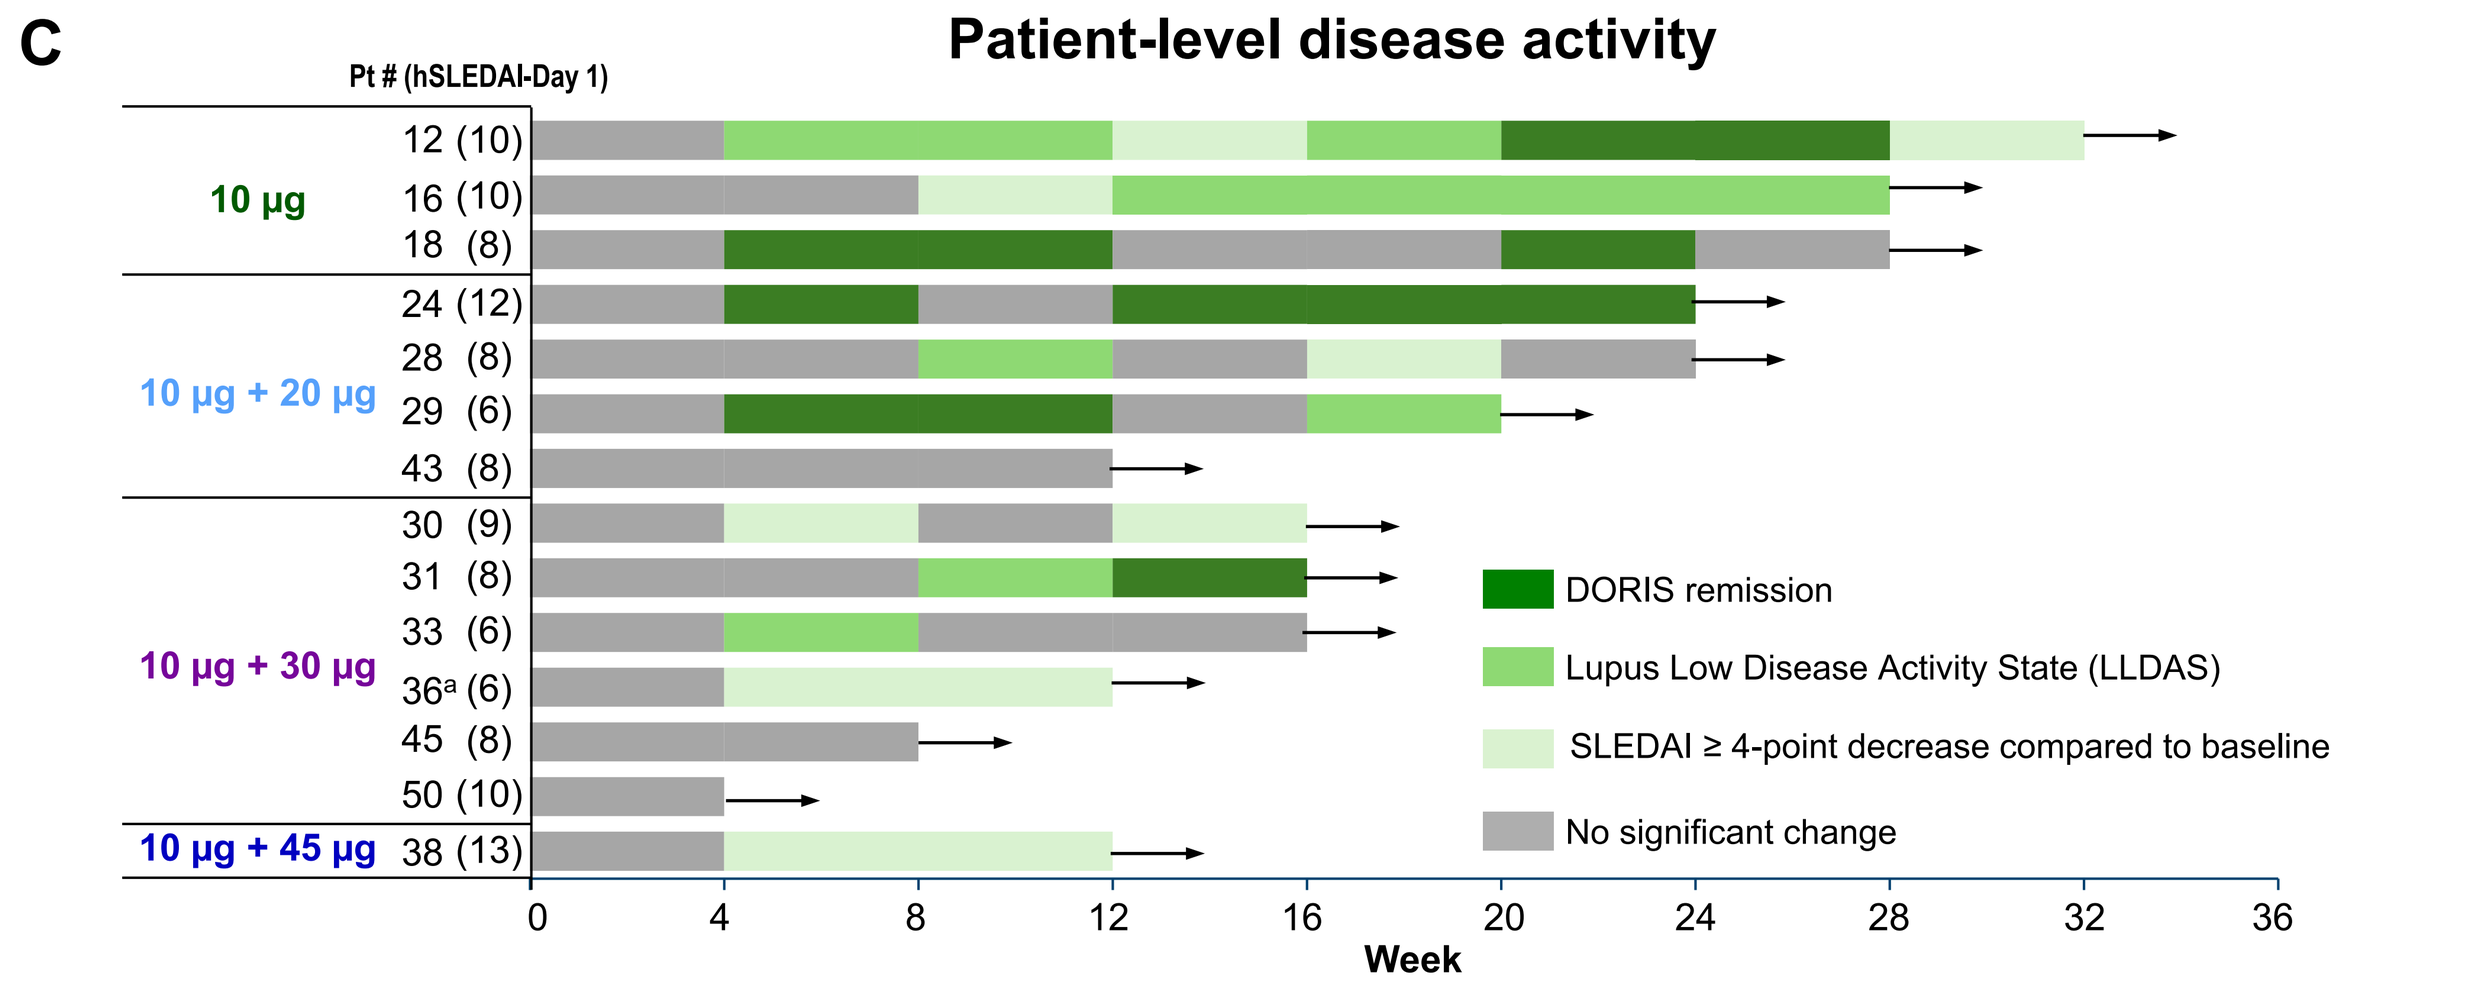

C
Patient-level disease activity
Pt # (hSLEDAI-Day 1)
10 µg
12 (10)
16 (10)
18 (8)
10 µg + 20 µg
24 (12)
28 (8)
29 (6)
43 (8)
10 µg + 30 µg
30 (9)
31 (8)
33 (6)
36ª (6)
45 (8)
50 (10)
10 µg + 45 µg
38 (13)
DORIS remission
Lupus Low Disease Activity State (LLDAS)
SLEDAI ≥ 4-point decrease compared to baseline
No significant change
Week

ª Only 1 patient (#36) had increase in SLE background therapy; prednisone increased at Week 3, and MMF initiated at Week 8 for ongoing pleuritic pain.

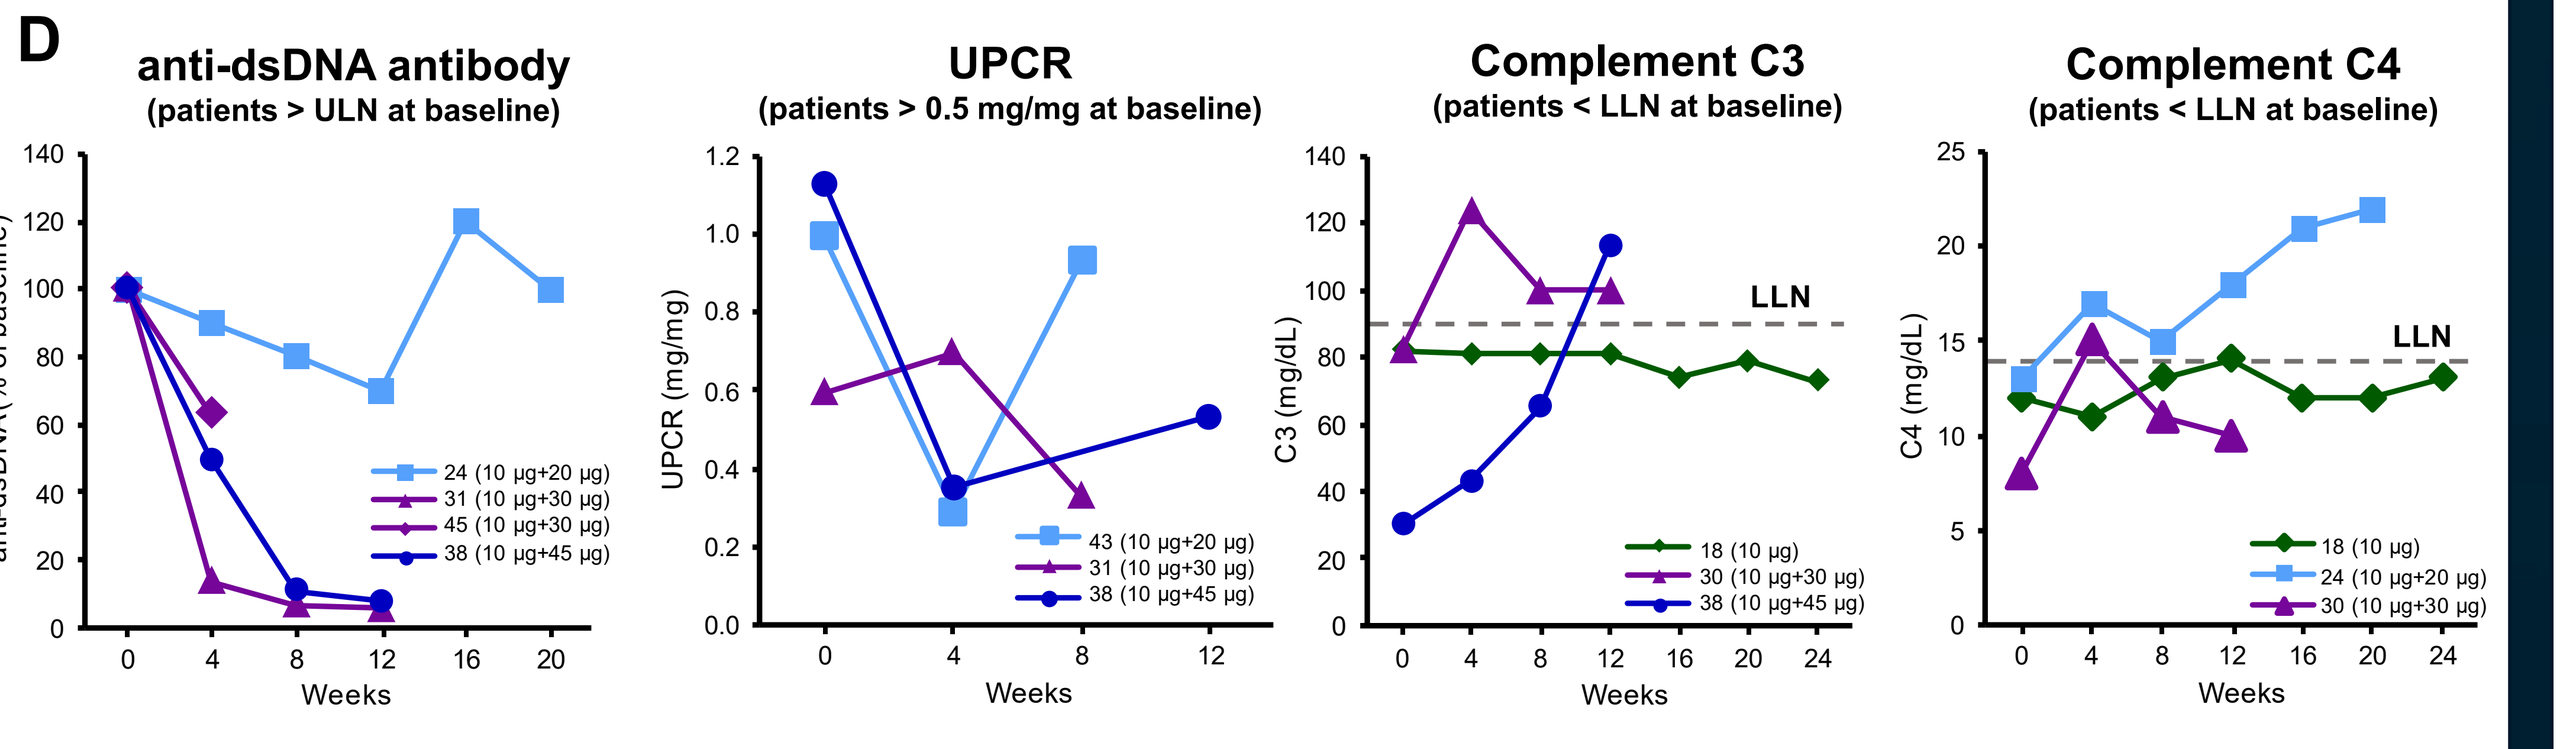

D
anti-dsDNA antibody
(patients > ULN at baseline)
anti-dsDNA (% of baseline)
24 (10 µg+20 µg)
31 (10 µg+30 µg)
45 (10 µg+30 µg)
38 (10 µg+45 µg)
UPCR
(patients > 0.5 mg/mg at baseline)
UPCR (mg/mg)
43 (10 µg+20 µg)
31 (10 µg+30 µg)
38 (10 µg+45 µg)
Complement C3
(patients < LLN at baseline)
C3 (mg/dL)
18 (10 µg)
30 (10 µg+30 µg)
38 (10 µg+45 µg)
Complement C4
(patients < LLN at baseline)
C4 (mg/dL)
18 (10 µg)
24 (10 µg+20 µg)
30 (10 µg+30 µg)
LLN
Weeks

Anti-dsDNA antibody titers, UPCR, C3, and C4 in patients on-treatment with CLN-978 shown as % from Baseline (anti-dsDNA abs) or concentration (mg/mL (dL)) for several patients treated in Cohorts 1-4.
Anti-dsDNA decreased to below ULN in patients 31 and 38, starting from 17 X ULN and 11 X ULN, respectively.
LLN=lower limit of normal; ULN=upper limit of normal.

Rheumatoid Arthritis

- Peripheral B-cell depletion BLOQ was achieved in 4/6 RA patients treated at target doses ≥ 20 µg (Cohorts 2 and 3) **(A)**
- Among the 7 patients with ≥ 12 weeks of data in this heavily pretreated population, 6 had high baseline disease activity
- Disease activity improved in 5 of 7 patients, including DAS28-ESR remission in 1 patient treated with a single 30 µg target dose (Cohort 3) **(C)**
 - 30 µg target dose showed greater improvements in ultrasound-assessed synovitis
- CLN-978 reduced RA autoantibody levels without impact on protective vaccine titers **(D)**

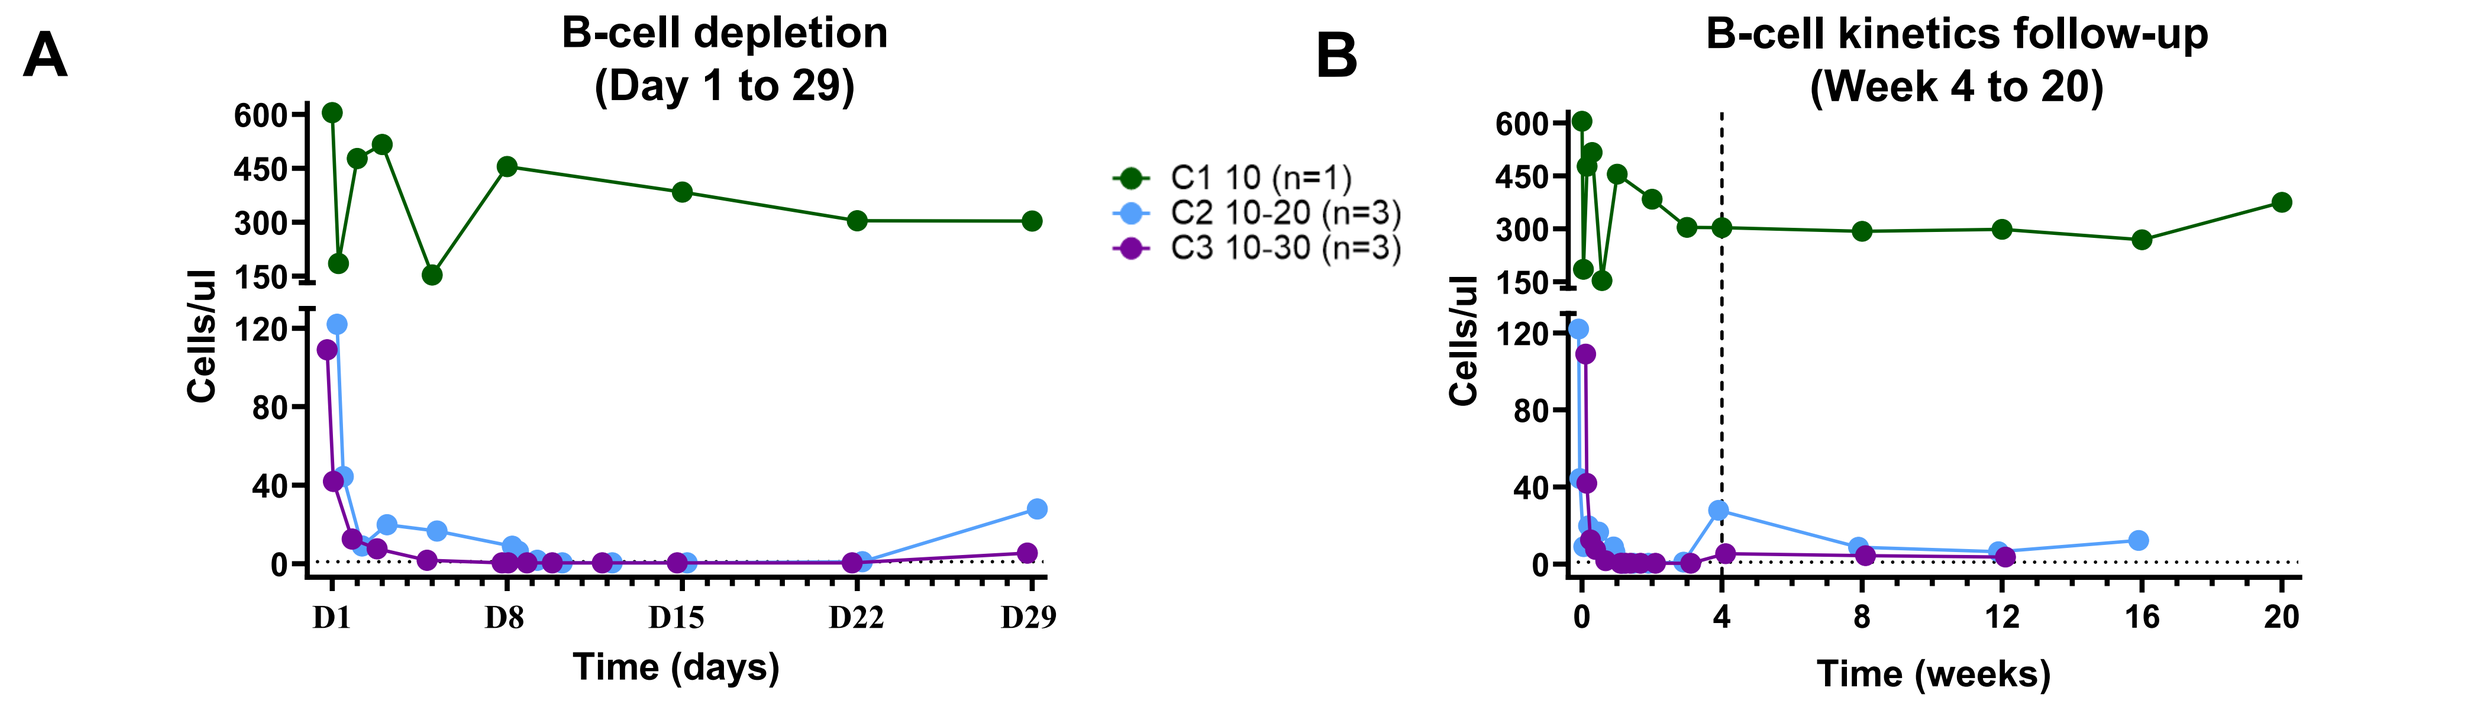

A
B-cell depletion (Day 1 to 29)
C1 10 (n=1)
C2 10-20 (n=3)
C3 10-30 (n=3)
Cells/ul
Time (days)
B
B-cell kinetics follow-up (Week 4 to 20)
Time (weeks)

Reduction in B-cell numbers during Days 1-29 after start of treatment (A) and B-cell kinetics follow-up during weeks 4-20 (B), shown as median per dose level as indicated. B-cell kinetics was measured by a validated TBNK assay with LLOQ of 0.9 cells/µL.

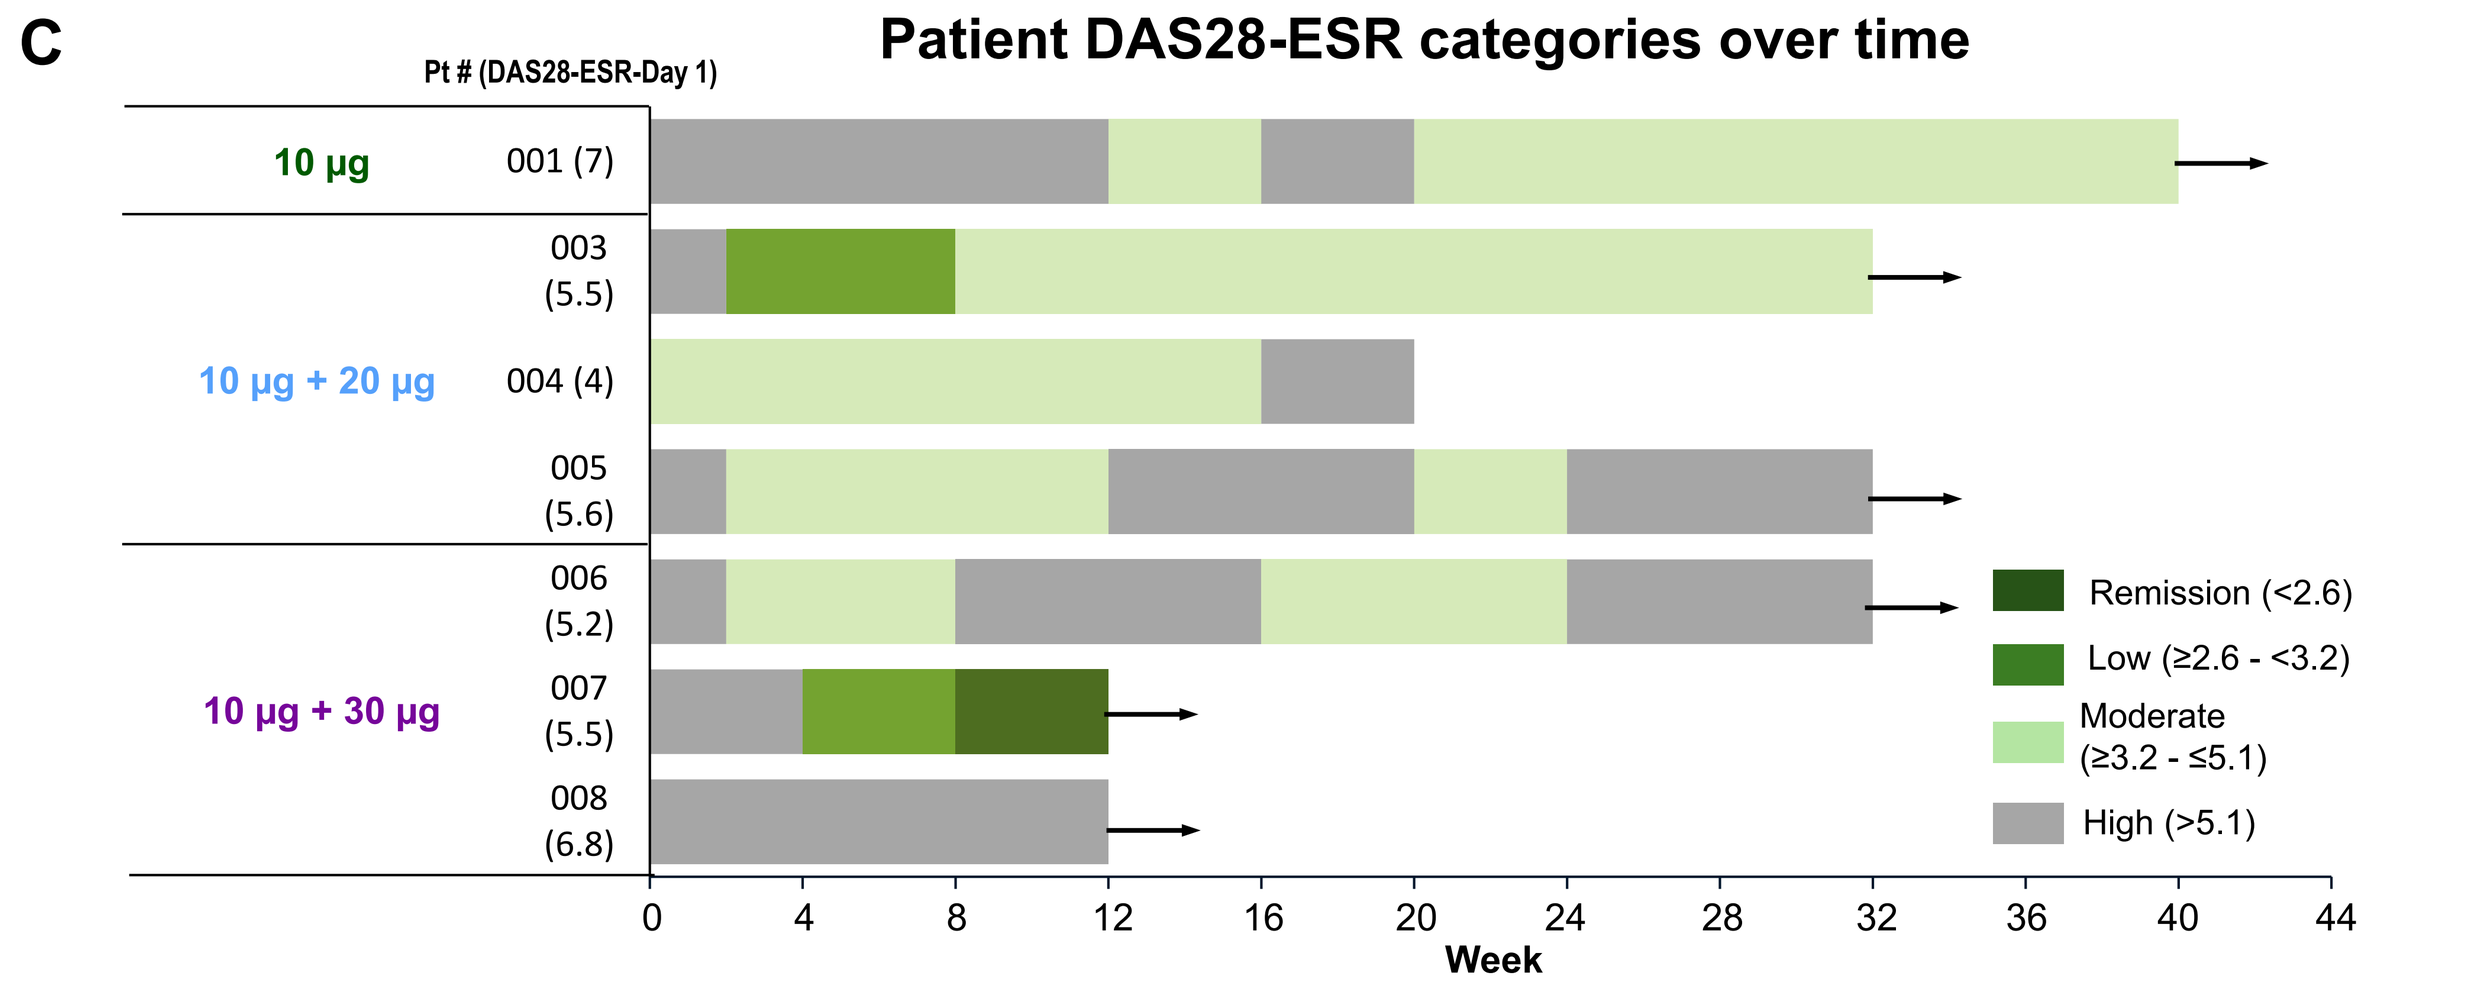

C
Patient DAS28-ESR categories over time
Pt # (DAS28-ESR-Day 1)
10 µg
001 (7)
10 µg + 20 µg
003 (5.5)
004 (4)
005 (5.6)
10 µg + 30 µg
006 (5.2)
007 (5.5)
008 (6.8)
Remission (<2.6)
Low (≥2.6 - <3.2)
Moderate (≥3.2 - ≤5.1)
High (>5.1)
Week

Three patients (01, 06, and 08) initiated ≥ 1 DMARD, on Days 58, 42, and 30, respectively.
C=cohort; D=day; DAS28-ESR=Disease Activity Score-28 with erythrocyte sedimentation rate.

D Reduction in autoantibody levels > 50% of baseline were observed in most patients treated with 20 µg or 30 µg target doses (Cohorts 2 and 3)

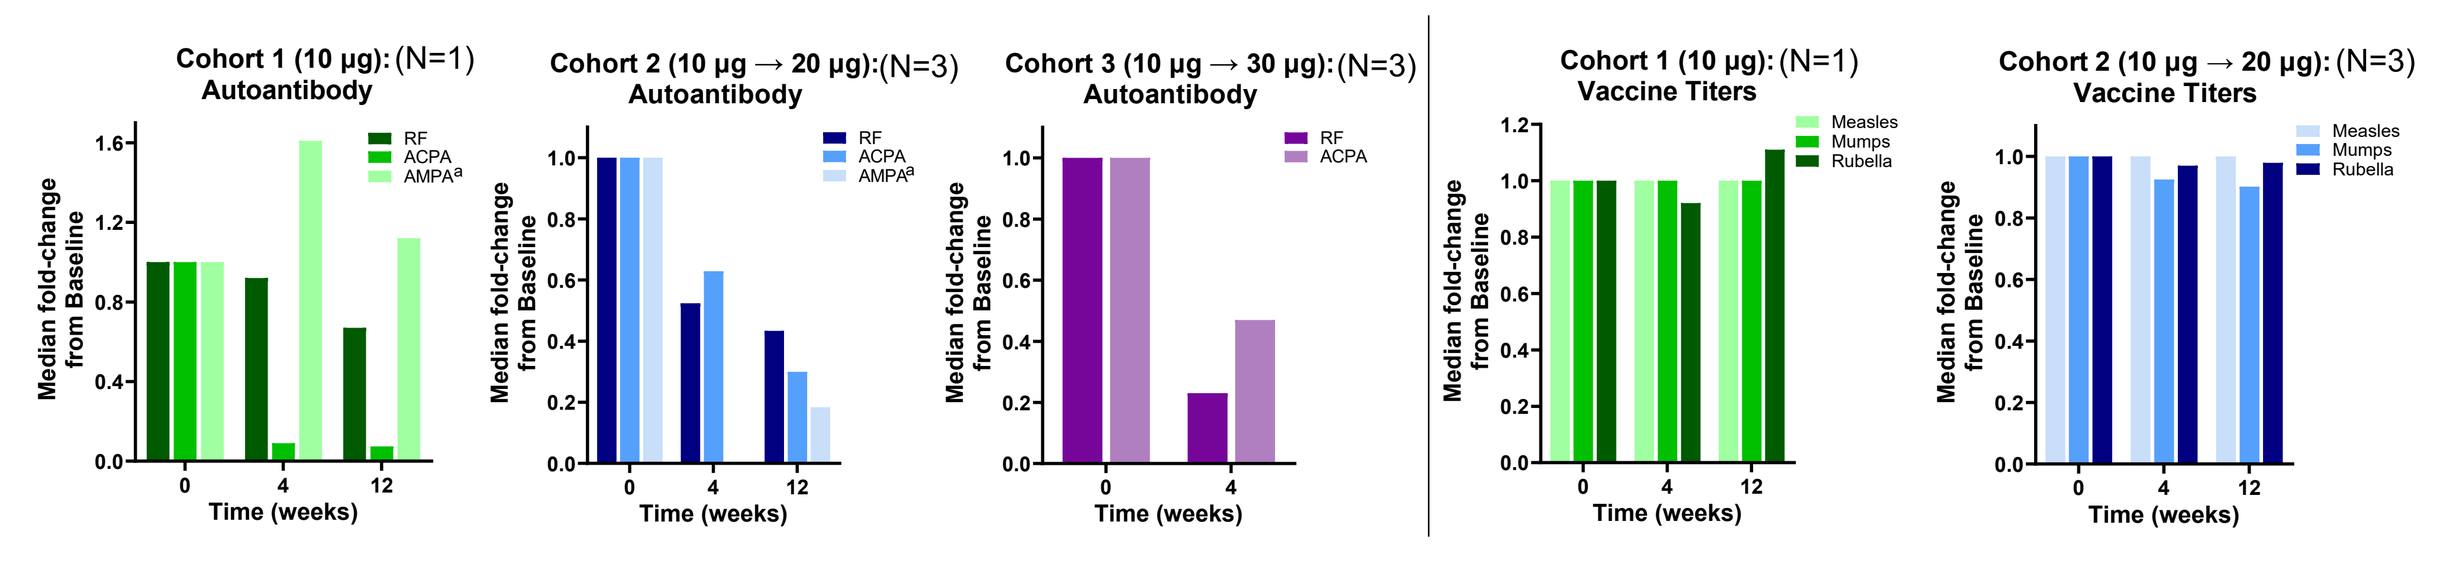

Cohort 1 (10 µg): (N=1) Autoantibody
Cohort 2 (10 µg → 20 µg): (N=3) Autoantibody
Cohort 3 (10 µg → 30 µg): (N=3) Autoantibody
Cohort 1 (10 µg): (N=1) Vaccine Titers
Cohort 2 (10 µg → 20 µg): (N=3) Vaccine Titers
RF
ACPA
AMPAª
Measles
Mumps
Rubella
Median fold-change from Baseline
Time (weeks)

Graphs depict median fold change from baseline for RF, ACPA and AMPA autoantibodies and measles, mumps, rubella vaccine titers.
ª AMPA was measured in two patients on study.
ACPA=anti-citrullinated protein antibodies; AMPA=anti-modified peptide antibodies; RA=rheumatoid arthritis; RF=rheumatoid factor.

RA CASE STUDIES: DOSE-DEPENDENT B-CELL DEPLETION IN THE TISSUE

- Peripheral B-cell depletion is necessary but not sufficient for tissue remodeling
- 20 µg target dose of CLN-978 effectively depleted B-cells at the periphery and in the synovium
- 30 µg target dose of CLN-978 disrupted follicular architecture in the lymph node and was accompanied by DAS28-ESR remission

A Peripheral B-cell depletion observed after 20 µg and 30 µg target doses of CLN-978

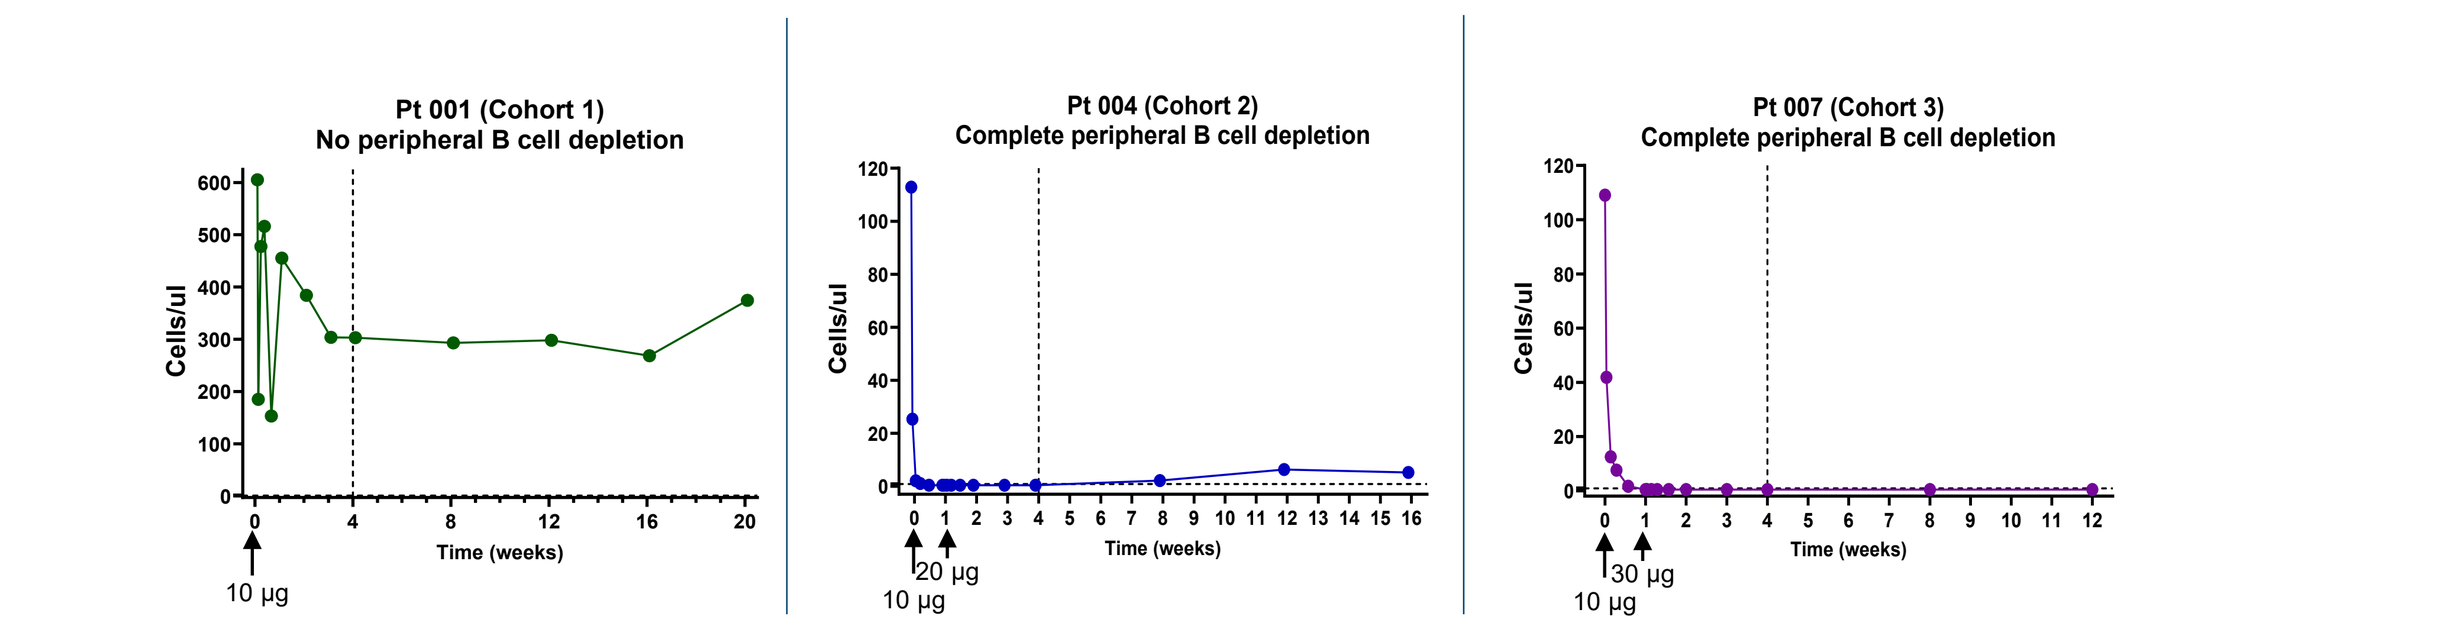

Pt 001 (Cohort 1)
No peripheral B cell depletion
Pt 004 (Cohort 2)
Complete peripheral B cell depletion
Pt 007 (Cohort 3)
Complete peripheral B cell depletion
Cells/ul
Time (weeks)
10 µg
20 µg
30 µg

B 20 µg and 30 µg target doses of CLN-978 caused B-cell depletion in the synovium

Post-treatment synovial biopsy (Day 29)

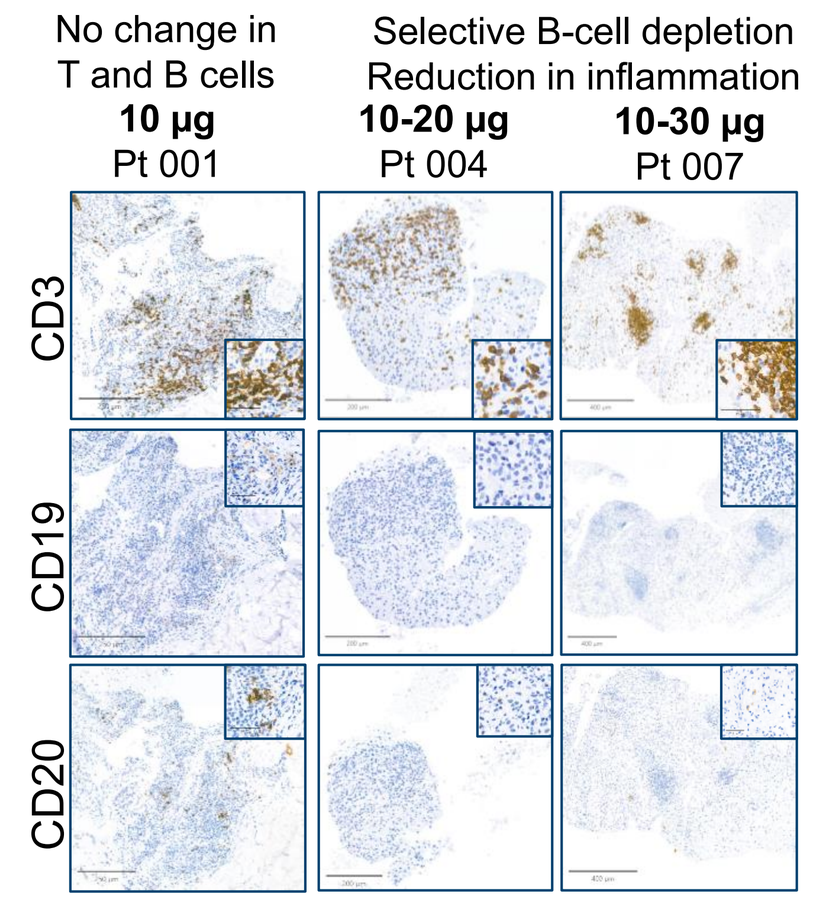

No change in T and B cells
10 µg
Pt 001
Selective B-cell depletion
Reduction in inflammation
10-20 µg
Pt 004
10-30 µg
Pt 007
CD3
CD19
CD20

Representative immunohistochemistry (IHC) images of patient's post-treatment (Day 29) synovium biopsies.

Change in immune cell density in the pre-treatment (screening) vs post-treatment synovial biopsy (Day 29)

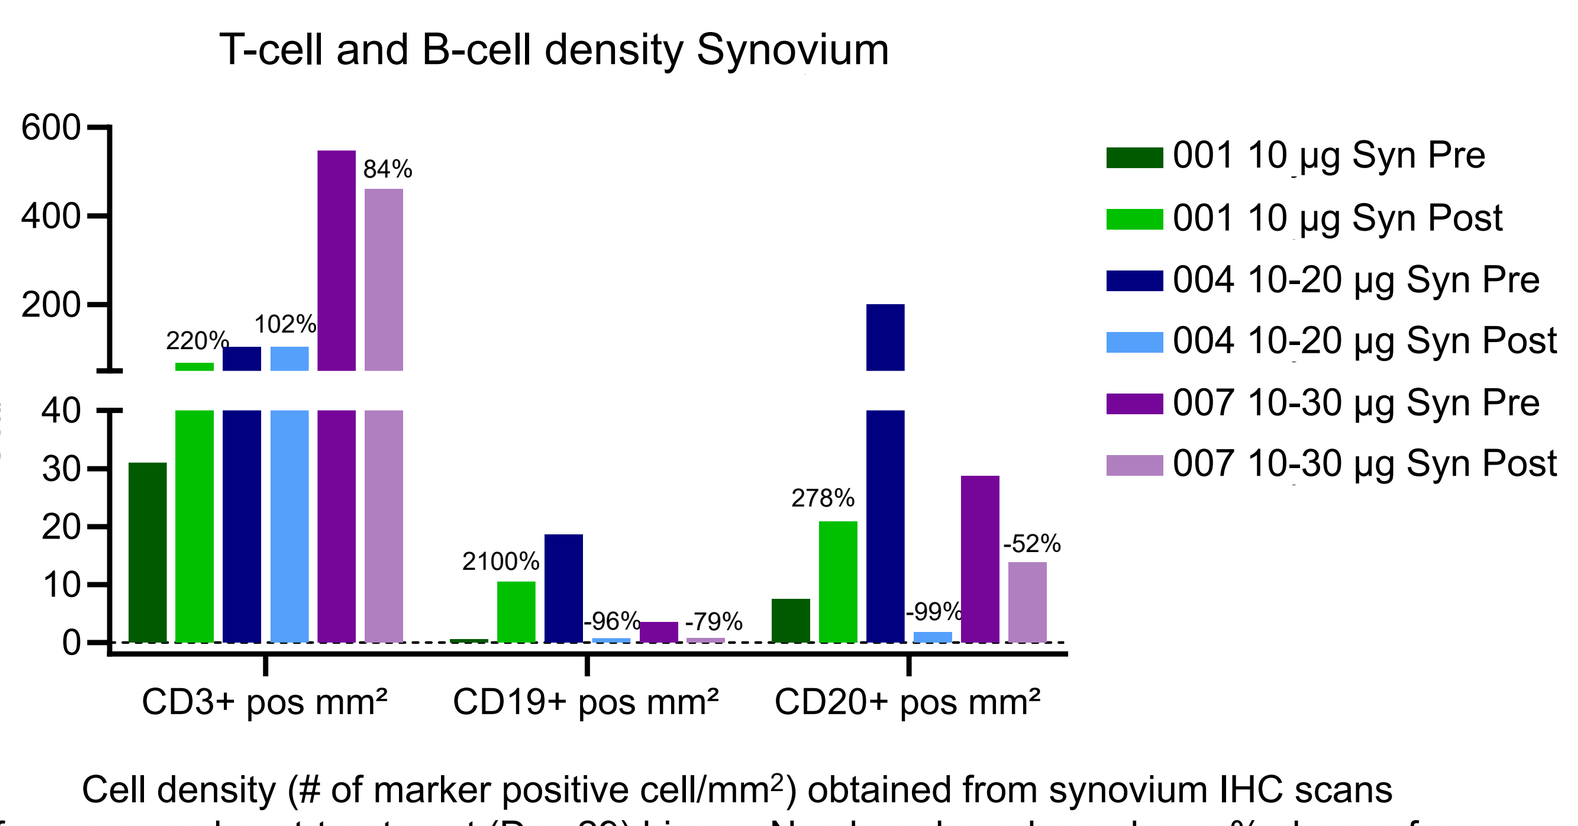

T-cell and B-cell density Synovium
Cell/mm2
220%
102%
84%
2100%
-96%
-79%
278%
-99%
-52%
CD3+ pos mm²
CD19+ pos mm²
CD20+ pos mm²
001 10 µg Syn Pre
001 10 µg Syn Post
004 10-20 µg Syn Pre
004 10-20 µg Syn Post
007 10-30 µg Syn Pre
007 10-30 µg Syn Post

Cell density (# of marker positive cell/mm^2) obtained from synovium IHC scans pre- and post-treatment (Day 29) biopsy. Number above bars shows % change from baseline (cells/mm^2 post (Day 29) / cells/mm^2 pre-treatment).

C 30 µg target dose of CLN-978 caused depletion of B-cells in the lymph node and disruption of the follicular architecture

Post-treatment lymph node biopsy (Day 29)

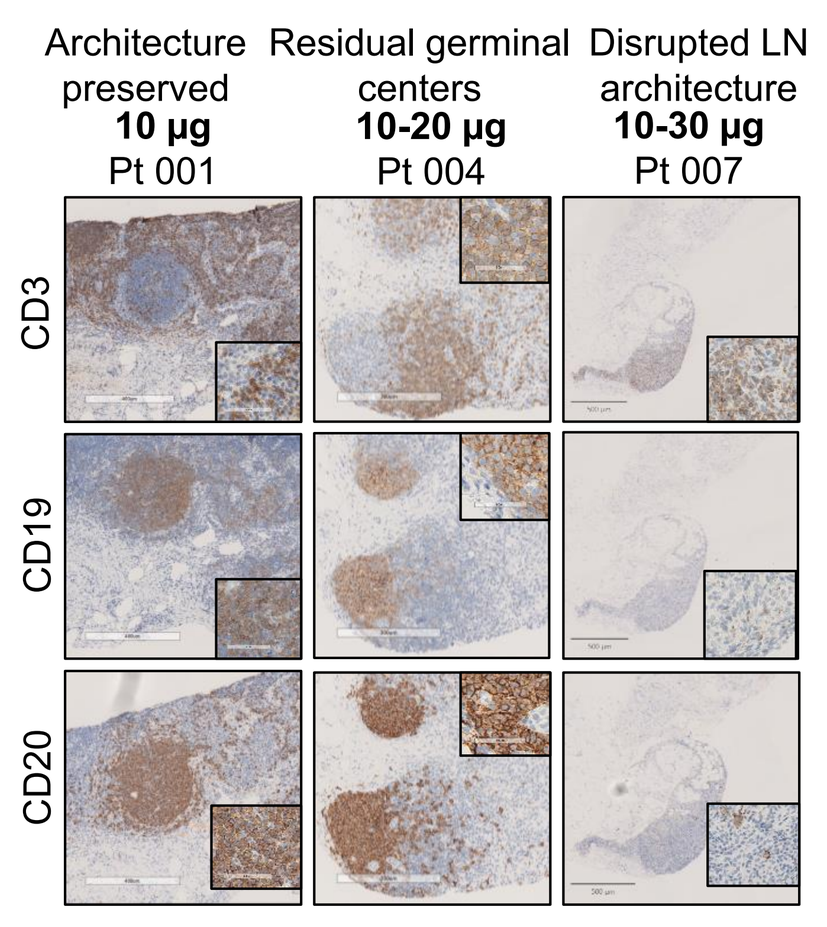

Architecture preserved
10 µg
Pt 001
Residual germinal centers
10-20 µg
Pt 004
Disrupted LN architecture
10-30 µg
Pt 007
CD3
CD19
CD20

Representative immunohistochemistry (IHC) images of patient's post-treatment (Day 29) LN biopsies.

Change in immune cell density in the pre-treatment (screening) vs post-treatment lymph node biopsy (Day 29)

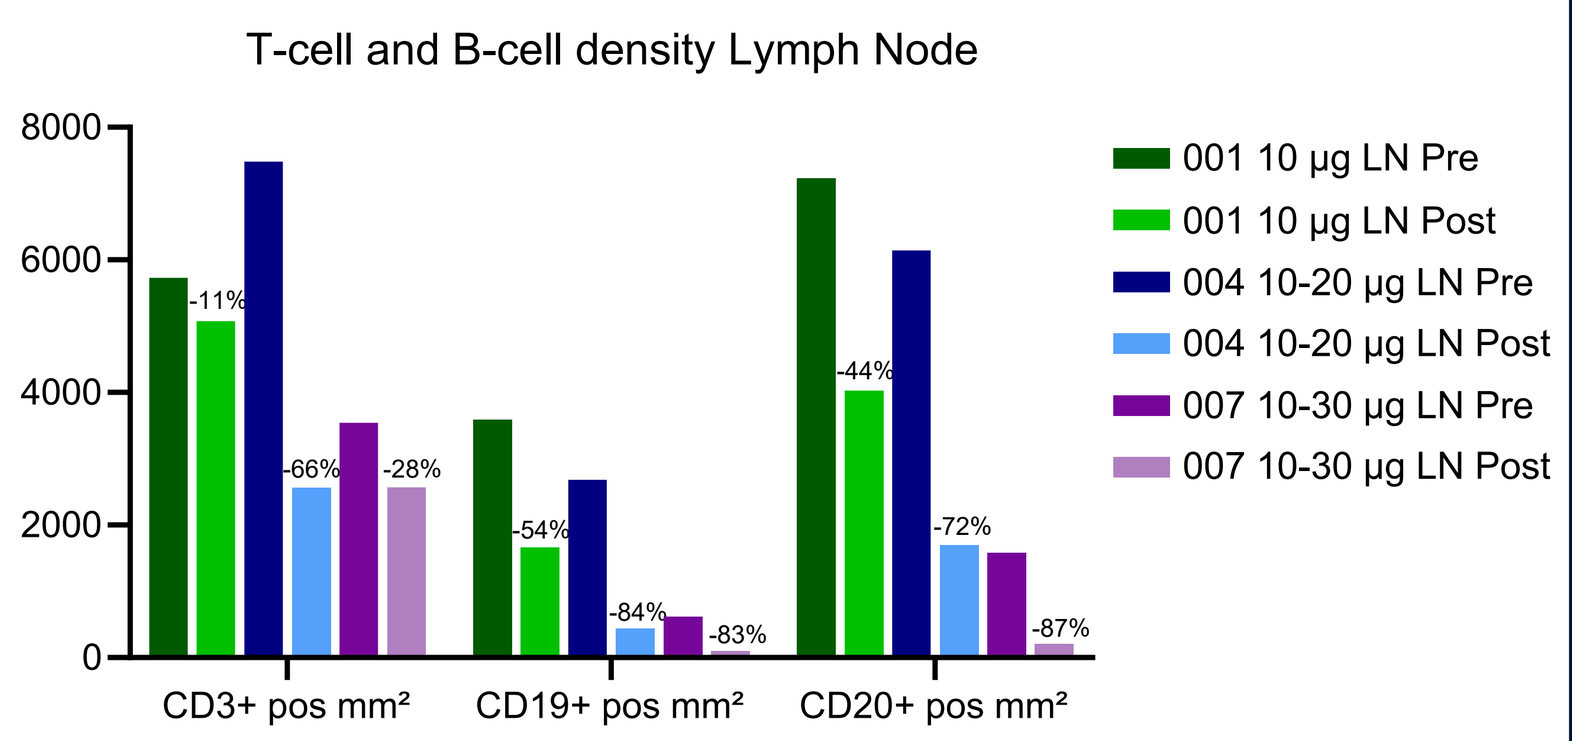

T-cell and B-cell density Lymph Node
Cell/mm2
-11%
-66%
-28%
-54%
-84%
-83%
-44%
-72%
-87%
CD3+ pos mm²
CD19+ pos mm²
CD20+ pos mm²
001 10 µg LN Pre
001 10 µg LN Post
004 10-20 µg LN Pre
004 10-20 µg LN Post
007 10-30 µg LN Pre
007 10-30 µg LN Post

Cell density (# of marker positive cell/mm^2) obtained from lymph node IHC scans pre- and post-treatment (Day 29) biopsy. Number above bars shows % change from baseline (# cells/mm^2 post (Day 29) / # cells/mm^2 pre-treatment).

D CLN-978 induced dose-dependent disruption of the lymph node follicular architecture especially pronounced after 30 µg target dose

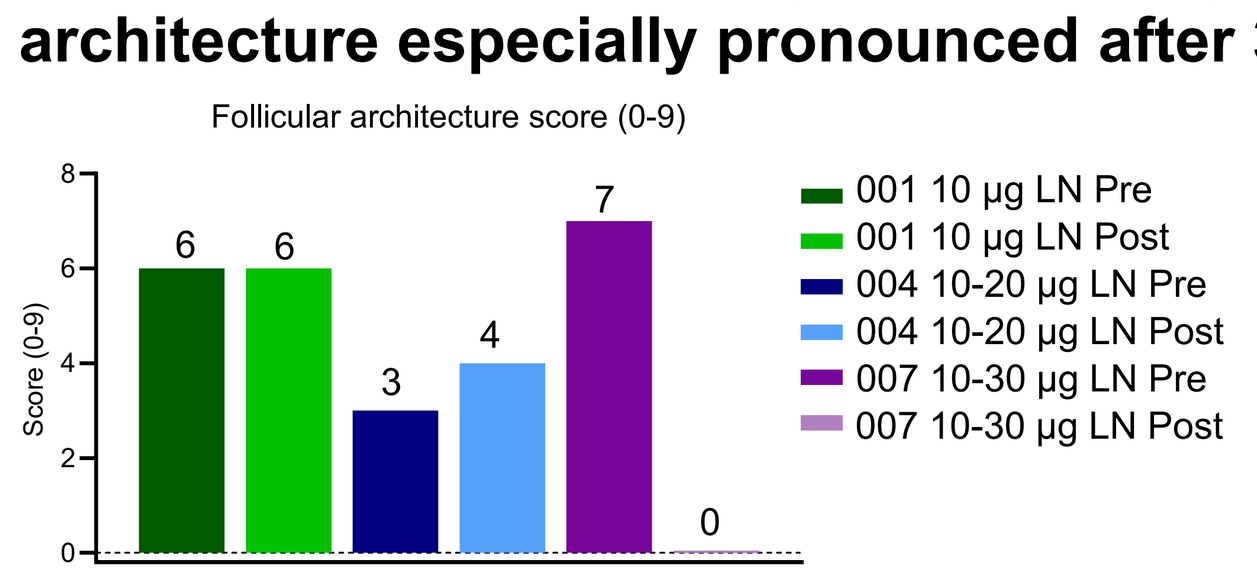

Follicular architecture score (0-9)
Score (0-9)
6
6
3
4
7
0
001 10 µg LN Pre
001 10 µg LN Post
004 10-20 µg LN Pre
004 10-20 µg LN Post
007 10-30 µg LN Pre
007 10-30 µg LN Post

Follicular architecture score: a semiquantitative score integrating general distribution and presence of B cells, architecture of the FDC network (CD21), proliferation rate of germinal center B cells (Ki67), and presence of TFH cells (PD1).[6]

Score = 0, complete disruption

SAFETY PROFILE IN PATIENTS WITH RA AND SLE

- CLN-978 was well tolerated across the 20 µg and 30 µg target dose cohorts, including patients who received multiple doses of 20 µg (Cohort 5)
- Most cytokine release syndrome (CRS) events were Grade 1 and occurred following the first dose (10 µg). A single case of Grade 3 CRS was observed following administration of the 45-µg target dose; enrolment to this cohort was discontinued, and additional step-up dosing is being implemented in multiple dose regimens
- No immune effector cell–associated neurotoxicity syndrome (ICANS) was observed

N = 29	Any grade n (%)	Grade ≥ 3 n (%)
Number of Subjects with at least 1 TEAE	24 (82.8)	5 (17.2)
Serious TEAE [a]	3 (10.3)	
Any TEAEs occurring in ≥ 3 patients:		
Cytokine release syndrome (CRS)	11 (37.9)	1 (3.4)
Headache	7 (24.1)	0
Fatigue	6 (20.7)	0
Diarrhea	4 (13.8)	0
Hypokalemia	4 (13.8)	0
Tachycardia	4 (13.8)	0
Alanine aminotransferase increased	3 (10.3)	0
Body temperature increased [b]	3 (10.3)	0
Nasopharyngitis	3 (10.3)	0
Urinary tract infection	3 (10.3)	1 (3.4)

CRS, n	Cohort 1 (n = 4) D1: 10 µg	Cohort 2 (n = 10) D1: 10 µg D8: 20 µg	Cohort 3 (n = 10) D1: 10 µg D8: 30 µg	Cohort 4 (n = 1) D1: 10 µg D8: 45 µg	Cohort 5 (n = 4) D1: 10 µg D8/15/22: 20 µg
Grade 1	0	5	2	0	2
Grade 2	0	0	1	0	0
Grade 3	0	0	0	1	0
Grade 4	0	0	0	0	0

[a] Serious TEAEs included CRS (1 patient Grade 2, 1 patient Grade 3), and 1 patient ~23 weeks after 30 µg dose had UTI, hyperglycemia, and seizure (all 3 AEs considered unrelated).
[b] Elevated temperature < 38°C.
TEAE=treatment-emergent adverse event.

CONCLUSIONS

Data from the initial dose escalation cohorts in RA and SLE patients demonstrates that a single target dose of ≥ 20 µg results in:

- **Deep B-cell depletion, including blood and tissues**
- **Clinically meaningful improvement in both RA and SLE disease activity, including clinical remissions**
- **Clinically significant autoantibody decreases**
- **Favorable safety profile with single target doses up to 30 µg as well as initial multi-target dose regimen; multi-dose exploration with the 20 µg and 30 µg doses continues**

REFERENCES
1. Müller F, et al. *N Engl J Med.* 2024;390(8):687–700. 2. Li Y, et al. *Cell Res.* 2025;35(3):220–223. 3. Michaelson JS, Baeuerle PA. *J Exp Med.* 2024;221(5):e20240499. 4. Shah K, et al. *Clin Exp Immunol.* 2024;217(1):15–30. 5. Nagorsen D, Baeuerle PA. *Exp Cell Res.* 2011;317(9):1255–1260. 6. Tur C, et al. *Ann Rheum Dis.* 2025;84(12):2065-2074.

ACKNOWLEDGMENTS
The authors express sincere gratitude to all patients who chose to participate in this trial, as well as investigators who participated after this abstract submission: John Tesser, Pankhuri Gupta, Kathy Nicholls, Hanns-Martin Lorenz, Mihai Abobului, Daniela Fodor, and Elena Kartvelishvili. This study is sponsored by Cullinan Therapeutics, Inc. Medical writing support was provided by Peloton Advantage, LLC, an OPEN Health company.

DISCLOSURES
Ricardo Grieshaber-Bouyer: received speaker honoraria from AbbVie, Alfasigma, Bristol Myers Squibb, Cullinan, Eli Lilly, Gilead, Novartis, Sanofi, UCB, Xencor; grants from AstraZeneca, Boehringer Ingelheim Stiftung, Candid, Else Kröner-Fresenius-Stiftung, Kyverna, Lupus Research Alliance, Johnson & Johnson, Pfizer. Anca Dinu Askanase: investigator/Consultant with AbbVie, Amgen, AstraZeneca, Aurinia, Biogen, Bristol Myers Squibb, Idorsia, Janssen, Genentech, GSK, Mallinckrodt, Nkarta, Pfizer, Sanofi, UCB. Rob Will: none declared. Nedal Adi: none declared. Kwabena Ayesu: none declared. Adrian Reber, Yue Zhang, Sarai Faison, Todd Shearer, Irina Shapiro, Stephen Wax, Jeff Jones: hold stock/stock options in Cullinan Therapeutics, Inc.; employees of Cullinan Therapeutics, Inc. Maria Gabriella Raimondo: none declared. Carlo Tur: none declared. Stefano Alivernini: none declared. Georg Schett: received speaker honoraria from Novartis, Johnson & Johnson, Bristol Myers Squibb, UCB. Maria Antonietta D'Agostino received speaker honoraria from AbbVie, AstraZeneca, Amgen, Bristol Myers Squibb, Eli Lilly, Johnson & Johnson, Novartis, UCB.

Email for questions or comments: swax@cullinantx.com